Abstract# TPS3169 *TRIAL IN PROGRESS:* A Phase 1a/b Open Label Study of IK-175, an Oral AHR Inhibitor, Alone and in Combination with Nivolumab in Patients with Locally Advanced or Metastatic Solid Tumors and Urothelial Carcinoma

Meredith A. McKean[1], David H. Aggen[2], Nehal J. Lakhani[3], Babar Bashir[4,5], Jason J. Luke[6], Jean Hoffman-Censits[7], Omar Alhalabi[8], I. Alex Bowman[9], Elizabeth A. Guancial[10], Alan Tan[11], Trupti Lingaraj[12], Marissa Timothy[12], Marta Sanchez-Martin[12], Katherine Kacena[12], Karim S. Malek[12], and Sergio Santillana[12]

[1]Sarah Cannon Research Institute at Tennessee Oncology LLC, Nashville, TN, [2]Memorial Sloan Kettering Cancer Center, NY, NY, [3]START Midwest, Grand Rapids, MI, [4]Thomas Jefferson University, Philadelphia, [5]Sarah Cannon Research Institute, Nashville, TN, [6]University of Pittsburgh, Pittsburgh, PA, [7]Johns Hopkins, Baltimore, MD, [8]MD Anderson Cancer Center, Houston, TX, [9]Banner Health, Gilbert, AZ, [10]Florida Cancer Specialists, Sarasota, FL, [11]Rush University, Chicago, IL, [12]Ikena Oncology, Boston, MA

BACKGROUND

- Aryl Hydrocarbon Receptor (AHR) is a ligand-activated transcription factor that regulates the activity of multiple innate and adaptive immune cells
- AHR binds to several endogenous and exogenous ligands, including kynurenine, generated from the precursor tryptophan by IDO1 and TDO2 and leads to a net immunosuppressive tumor microenvironment, making AHR an attractive therapeutic target in multiple cancer types.
- IK-175 was selected based on mechanism of action and robust antitumor activity in preclinical models
- AHR up-regulates PD-1 on CD8+ T cells and therefore IK-175 may overcome the immunosuppressive effects driving resistance to nivolumab in certain subsets of cancer patients
- The combination of IK-175 and nivolumab was selected based on complementary mechanisms of action and robust antitumor activity in preclinical models.

IK-175 is a selective, small molecule AHR inhibitor being developed as an oral (PO) agent. In human T-cells, IK-175 induces an activated T-cell state, interleukin (IL)-22 gene expression, and leads to an increase in proinflammatory cytokines, such as IL-2 and IL-9.

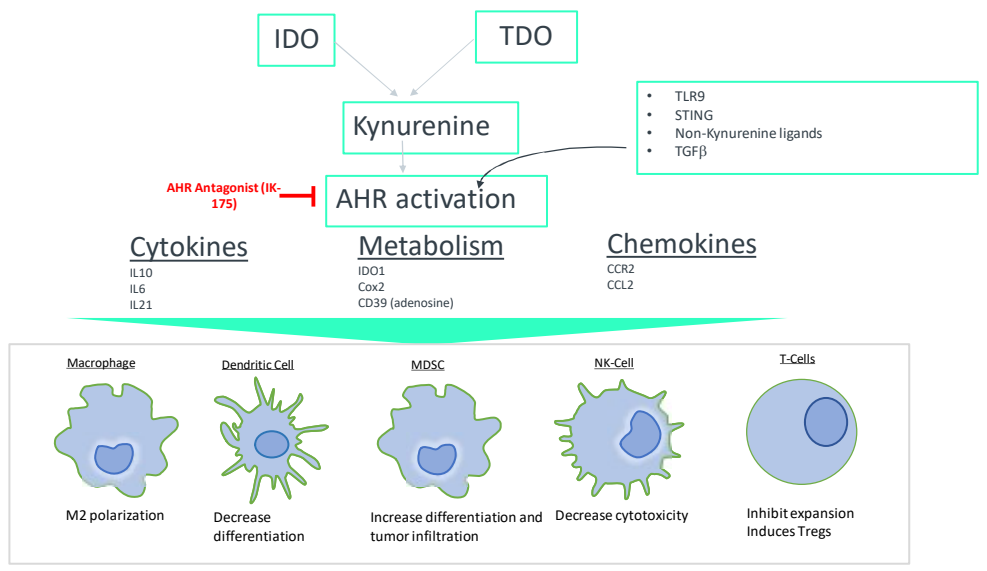
IDO
TDO
Kynurenine
TLR9
STING
Non-Kynurenine ligands
TGFβ
AHR Antagonist (IK-175)
AHR activation
Cytokines
IL10
IL6
IL21
Metabolism
IDO1
Cox2
CD39 (adenosine)
Chemokines
CCR2
CCL2
Macrophage
Dendritic Cell
MDSC
NK-Cell
T-Cells
M2 polarization
Decrease differentiation
Increase differentiation and tumor infiltration
Decrease cytotoxicity
Inhibit expansion Induces Tregs

RATIONALE

- Transcriptional analysis of TCGA data using a proprietary AHR gene signature demonstrated high expression of AHR and AHR signature genes in bladder cancer
- Genomic profiling of cancers from TCGA and Project GENIE demonstrated the highest prevalence of AHR amplification in bladder cancer
- AHR immunohistochemistry (IHC) tumor microarray analysis across 15 different tumor types revealed that bladder cancer has the highest level of AHR protein expression and AHR nuclear localization, an indicator of active AHR signaling
- Therefore, nuclear AHR in urothelial carcinoma tumors is being investigated for potential predictive clinical benefit with IK-175

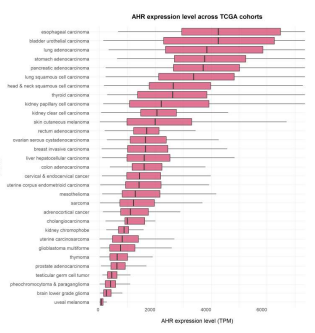
AHR expression level across TCGA cohorts

AHR Target Gene Expression

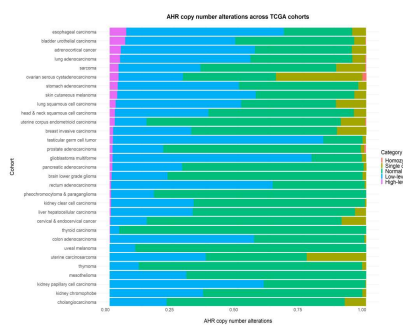
AHR copy number alterations across TCGA cohorts

AHR amplification

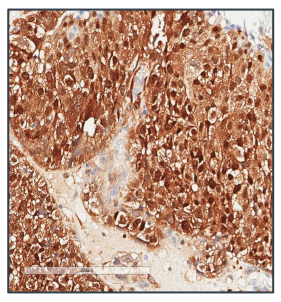

IHC

METHODS

Study Design

- This is a first-in-human (FIH), multicenter, open-label, phase 1a/b dose escalation and expansion study assessing the safety and preliminary antitumor activity of IK-175, an AHR inhibitor, as a single agent and in combination with nivolumab in patients with advanced solid tumors and urothelial carcinoma
 - Dose escalation will enroll patients with solid tumors and will include a minimum of 3 subjects at each dose level
 - Dose expansion will enroll patients with urothelial carcinoma patients only in both the single agent and combination arms at the selected expansion dose
- Enrichment by prospective assessment for AHR nuclear localization by IHC in patients with urothelial carcinoma in expansion cohorts of both treatment arms
 - A minimum of 10 patients having a positive AHR nuclear localization test (cutoff for positive AHR is 65% tumor cells positive for 2+/3+ nuclear AHR by a validated IHC assay) will be enrolled in the combination arm

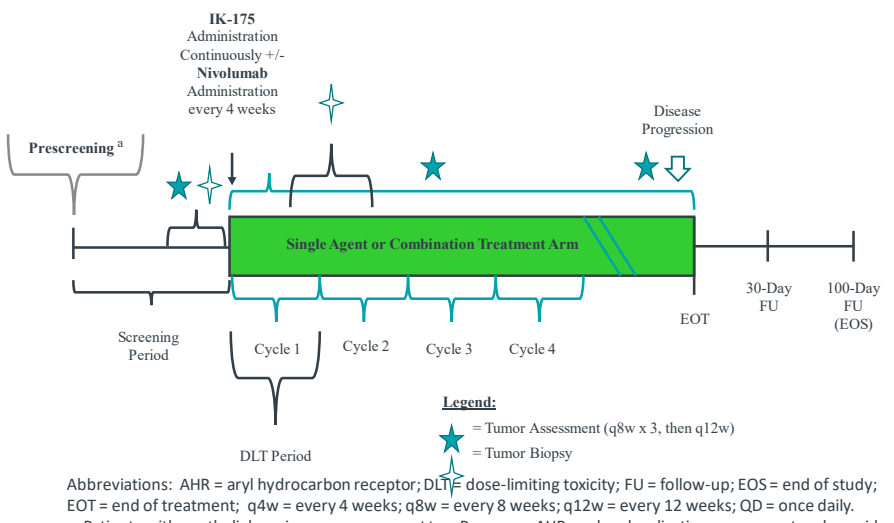
IK-175 Administration Continuously +/- Nivolumab Administration every 4 weeks
Prescreening a
Disease Progression
Single Agent or Combination Treatment Arm
Screening Period
Cycle 1
Cycle 2
Cycle 3
Cycle 4
DLT Period
EOT
30-Day FU
100-Day FU (EOS)
Legend:
= Tumor Assessment (q8w x 3, then q12w)
= Tumor Biopsy

Abbreviations: AHR = aryl hydrocarbon receptor; DLT = dose-limiting toxicity; FU = follow-up; EOS = end of study; EOT = end of treatment; q4w = every 4 weeks; q8w = every 8 weeks; q12w = every 12 weeks; QD = once daily.
a. Patients with urothelial carcinoma may consent to a Prescreen AHR nuclear localization assessment and provide archival tumor tissue prior to the Screening period. There is no time limit (i.e., window) for this assessment during the Prescreening period.

Dosing Schedule

	IK-175	Nivolumab[a]
Route of administration	Oral	IV
Phase 1a dose	Dose per dose escalation scheme[b]	480mg
Phase 1b dose	1200mg	480mg
Dosing schedule and period	QD in continuous 21[c] or 28-day cycles	Q4w on Day 1 of 28-day cycles

a. for combination arm only; b. 5 dose levels tested starting at 200mg QD; c. for single agent arm only

Study Outcomes

- Primary safety objectives are to determine the MTD or MAD, to evaluate DLT's, and to determine the RP2D of IK-175 as a single agent and in combination with nivolumab
- Key secondary efficacy and PK endpoints include ORR, DCR, DOR, DOT, PFS, $t_{1/2}$, AUC and Cmax
- Additional secondary endpoints for urothelial carcinoma patients include antitumor assessments per iRECIST

Key Eligibility Criteria

INCLUSION CRITERIA	EXCLUSION CRITERIA
Adult patients with histologically confirmed solid tumors who have locally recurrent or metastatic disease that has progressed on or following all standard of care therapies	Clinically unstable central nervous system (CNS) tumors or brain metastasis (stable and/or asymptomatic CNS metastases allowed)
Urothelial carcinoma patients in the combination arm must have progressed on or within 3 months of receiving the last infusion/dose of anti-PD-(L)1 therapy	Any condition requiring continuous systemic treatment with either corticosteroids (>10 mg daily prednisone equivalents) or other immunosuppressive medications within 2 weeks. Inhaled or topical steroids and physiological replacement doses are permitted in the absence of active clinically significant autoimmune disease
No limit to the number of prior treatment regimens. PD-(L)1 does not need to immediately precede trial	Active autoimmune disease that has required systemic treatment in past 2 years; nonsteroidal anti-inflammatory drugs (NSAIDs) are permitted
Have measurable disease per RECIST v1.1	Patients taking strong CYP3A4/5 inhibitors or inducers unless they can be transferred to other medications ≥ 5 half-lives prior to dosing

Statistical Considerations

- A target sample size of 26 patients with solid tumors for single agent and combination is planned for dose escalation and a target sample size of 67 patients with urothelial carcinoma for single agent and combination is planned for dose expansion
- A mTPI-2 design with a target DLT rate of 30% will be applied for dose escalation and to determine the expansion dose of IK-175 as a single agent and in combination with nivolumab
- A Simon 2-Stage design will be used in dose expansion to assess preliminary antitumor activity in patients with urothelial carcinoma in both treatment arms determined by ORR
- All safety analyses will be performed on the safety population defined as all subjects who received any amount of study treatment
- ORR will be analyzed using the response-evaluable population (defined as subjects who received at least 1 dose of study treatment, have measurable disease at baseline, and at least 1 post-baseline response assessment)

STUDY STATUS

The study started in January 2020 and is actively enrolling patients in dose expansion cohorts of both single agent and combination treatment arms. Clinical trial information: NCT04200963

REFERENCES

- McGovern K., Castro A., Cavanaugh J., Discovery of clinical candidate IK-175, a selective orally active AHR antagonist. SITC 2020, poster presentation
- Sanchez-Martin M, Wang L, McGovern K, Ecsedy J and Zhang M.: Computational biology and tissue-based approaches to inform indication selection for a novel AHR inhibitor. SITC 2021, poster presentation (#93)
- Wang L, Sanchez-Martin M, Tirrell S, Ortiz-Otero N and Zhang M.: Analytical validation of a novel immunohistochemistry assay to determine nuclear AHR expression in human bladder cancer. SITC 2021, poster presentation (#58)

ACKNOWLEDGEMENTS

- Ikena Oncology would like to recognize and express gratitude to the patients and their families who contributed to this study.