Poster #58

Analytical validation of a novel immunohistochemistry assay to determine nuclear AHR expression in human bladder cancer

Lei Wang[1] , Marta Sanchez-Martin[1], Steve Tirrell [1], Nerymar Ortiz-Otero[1], X. Michelle Zhang[1]
[1]Ikena Oncology, Inc. Boston, MA 02210

Background

- Aryl hydrocarbon receptor (AHR) is a ligand-activated transcription factor that translocates to the nucleus upon ligand binding, and activates a target gene expression program, contributing to the immunosuppressive state of the tumor microenvironment [1].
- IK-175 is a selective, oral, potent AHR antagonist that has demonstrated strong immune modulation, and robust tumor growth inhibition, as a single agent or in combination with anti-PD-1, in mouse models.
- IK-175 is being evaluated in an ongoing phase 1 clinical study as a single agent in advanced solid tumors and in combination with nivolumab for bladder cancer patients (NCT04200963).
- We hypothesized that nuclear AHR protein expression in tumors is an indicator of activated AHR signaling, and thus a potential predictive biomarker for the clinical response to IK-175.
- To test this hypothesis, we have developed a novel AHR immunohistochemistry (IHC) assay and analytically validated it in a Clinical Laboratory Improvement Amendments (CLIA) certified lab. We have successfully implemented it in our ongoing phase 1b clinical trial of IK-175 for prospective patient enrollment.

Work flow

Assay development
- Antibody screening and selection
- Staining protocol development using cell lines/xenografts
- In house and in collaboration with Flagship Biosciences (FLG)

Assay transfer and design
- Assay transfer and optimization in NeoGenomics (Neo)
- Concordance of nuclear AHR scoring between pathologist and computational analysis
- Cutoff value determination

Assay validation
- Accuracy, sensitivity and specificity
- Precision, intra-pathologist concordance
- Antigen stability

Implementation in clinical trial for patient enrichment

Assay development

Screening and selection of the optimal anti-AHR antibody

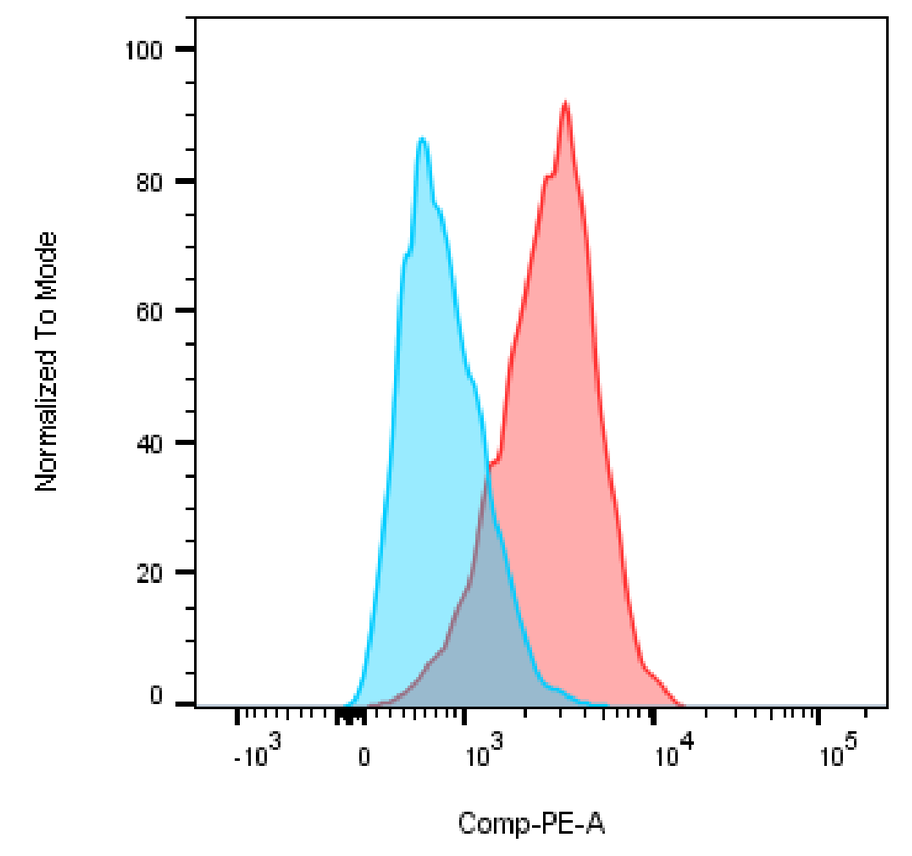
Normalized To Mode
100
80
60
40
20
0
-10^3
0
10^3
10^4
10^5
Comp-PE-A

Figure 1: anti-AHR antibody selection. Histogram showing AHR intensity in HeLa cells control siRNA treated cells vs AHR siRNA treated cells. Screening data not shown.

Red: control Blue: AHR siRNA treated cells

The developed assay showed broad dynamic range with well-distributed H scores of nuclear AHR expression

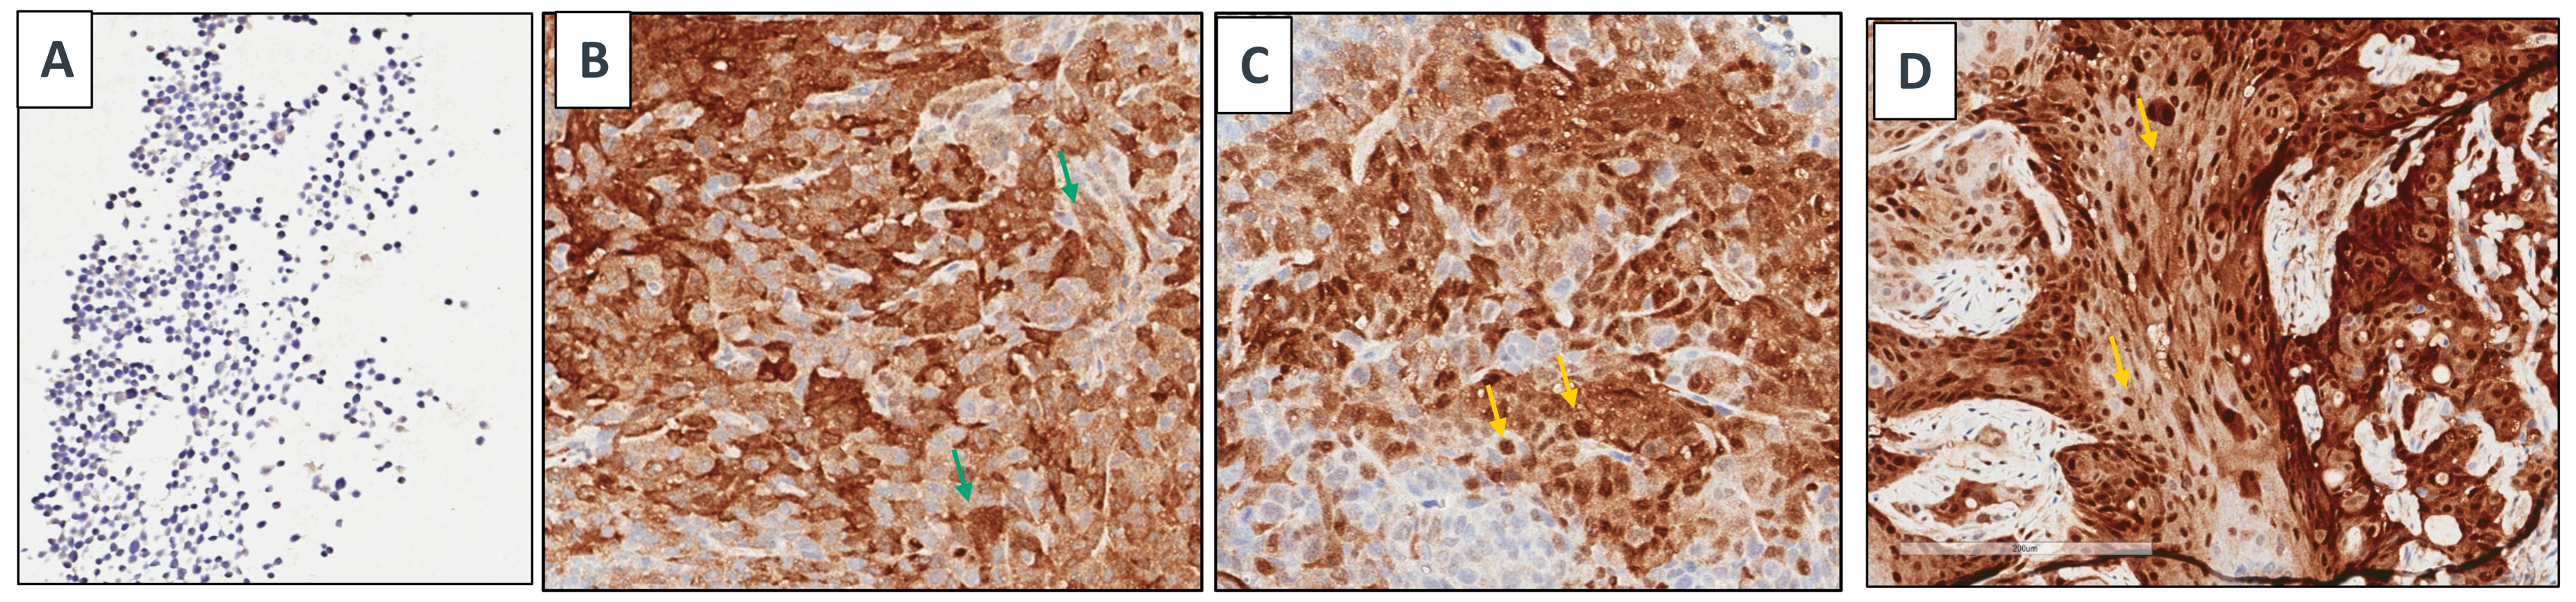
A
B
C
D

Figure 2: Assay development using Formalin Fixed Paraffin Embedded (FFPE) samples with well-characterized AHR expression. (A) Jurkat cell pellet, AHR nuclear negative (B) HCT-116 xenograft, low AHR nuclear expression (C) HCT-116 xenograft with AHR ligand treatment, median AHR nuclear expression, and (D) Cal-27 (AHR amplification) xenograft with AHR ligand treatment, high AHR nuclear expression. Green arrow: cytoplasmic staining. Yellow arrow: nuclear staining.

Assay transfer and optimization

Assay was successfully transferred from Leica Bond RX to a CDx-compatible Leica Bond III platform in Neo, and the optimized assay yielded more defined AHR nuclear staining and cleaner background (Fig. 3 C, D).

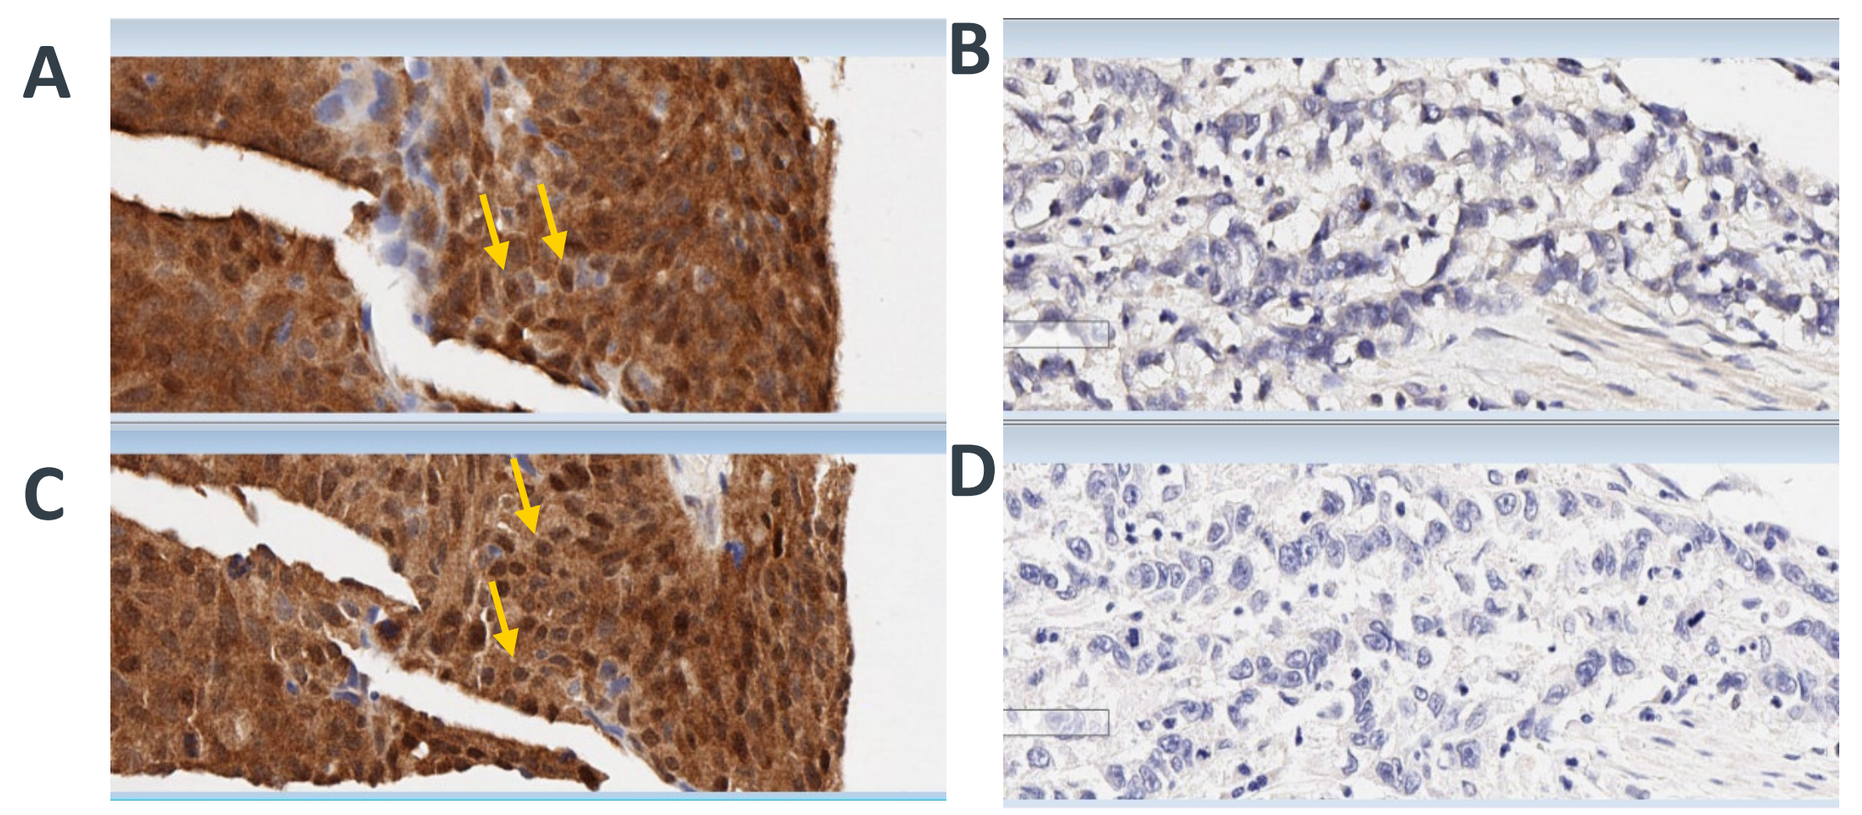
A
B
C
D

Figure 3: Assay transfer and optimization using human bladder cancer FFPE specimens. (A, B) staining images on Leica Bond RX platform (C, D) staining images on Leica Bond III platform.

Yellow arrow: nuclear staining.

Cutoff value determination

- Pathologist scoring was used as the path forward for potential CDx development.
- Staining intensity (0, 1+, 2+, or 3+) was determined for each cell
 - Cells in tumor region with an intensity of 2+ and 3+ were considered strong AHR nuclear positive
- N=110 human bladder cancer FFPE samples were used to establish a cutoff of nuclear 2+ and 3+ positive tumor ≥ 65% at Neo.
- This cutoff was set to capture the top 20-30% of expressers. Due to differences in observed overall staining intensity, a cutoff of 60% was set for the specimens stained at FLG, the reference laboratory.
- The cutoffs were then applied to a new independent set of 89 bladder cancer specimens. The staining results of these specimens from Neo were compared to those from FLG for the evaluation of accuracy, sensitivity, and specificity of the AHR IHC assay.

Assay validation- accuracy, sensitivity, and specificity

The assay accuracy, sensitivity, and specificity met the predefined acceptability criteria of ≥85%.

		AHR IHC - Reference (FLG)		
		Expected AHR Positive	Expected AHR Negative	Total
AHR IHC Staining (Neo)	Actual AHR Positive	15 (True Positives)	4 (False Positives)	21% (19/89)
	Actual AHR Negative	0 (False Negatives)	70 (True Negatives)	79% (70/89)
	Total	15	74	100% (89/89)

Acceptance criteria (≥85%)	
Accuracy	96%
Sensitivity	100%
Specificity	95%
Precision	100%

Adjacent sections from the 60 bladder cancer specimens plus the bladder cancer TMA were sent to Neo and FLG (as a secondary AHR IHC assay). Stained slides were evaluated by a Board-certified Neo pathologist using a manual brightfield microscope and the positive/ negative were determined by cutoff values.

Assay validation- precision, intra-pathologist concordance and antigen stability

The assay precision and intra-pathologist concordance met the predefined acceptability criteria.
Antigen stability: The variance is less than 15% between baseline and 4 weeks

	Results	Acceptability criteria
Precision: intra-operator	100%	≥85%
Precision: inter-operator	100%	≥85%
Intra-pathologist concordance	90%	≥90%
Antigen stability	4 weeks	variance of no more than 15% from baseline

- Precision repeatability: 10 specimens, 5 sections per specimen, run on same day
- Precision reproducibility: 10 specimens, 10 sections per specimen, run on 5 separate days with 2 sections each day
- Intra-pathologist concordance: one pathologist scored 20 samples at two different time points with 15 days wash-out period between scoring.
- Antigen stability: 3 specimens were sectioned at baseline (day 0) and stored at room temperature, the stored slides and freshly sectioned slides were stained at 2 weeks and 4 weeks time points.

Conclusion

A novel and robust nuclear AHR IHC assay for bladder cancer was developed and analytically validated in a CLIA certified lab and showed ≥95% accuracy, specificity, sensitivity, and precision, enabling its implementation in an IK-175 Ph1b clinical study (NCT04200963) for prospective patient enrichment.

References:
1, Campesato L., Budhu S., Tchaicha J., et al., Blockade of the AHR restricts a Treg-macrophage suppressive axis induced by L-Kynurenine. *Nature Communications.* (2020) 11:4011.

Correspondence to: Lei Wang, lwang@ikenaoncology.com; X. Michelle Zhang, mzhang@ikenaoncology.com