Computational Biology and Tissue-based Approaches to Inform Indication Selection for a Novel AHR Inhibitor

Marta Sanchez-Martin, Lei Wang, Karen McGovern, Jeffrey Ecsedy, X. Michelle Zhang
Ikena Oncology, Inc. Boston, MA 02210

Abstract #15472
Poster #93

Background: Aryl Hydrocarbon Receptor (AHR) is a ligand-activated transcription factor that regulates the activities of multiple innate and adaptive immune cell types. Various exogenous and endogenous ligands such as kynurenine bind to AHR and subsequently ARNT (AHR nuclear transportation) driving AHR nuclear translocation and transcriptional activation, leading to an immunosuppressive tumor microenvironment (Quintana 2013; Murray 2014).

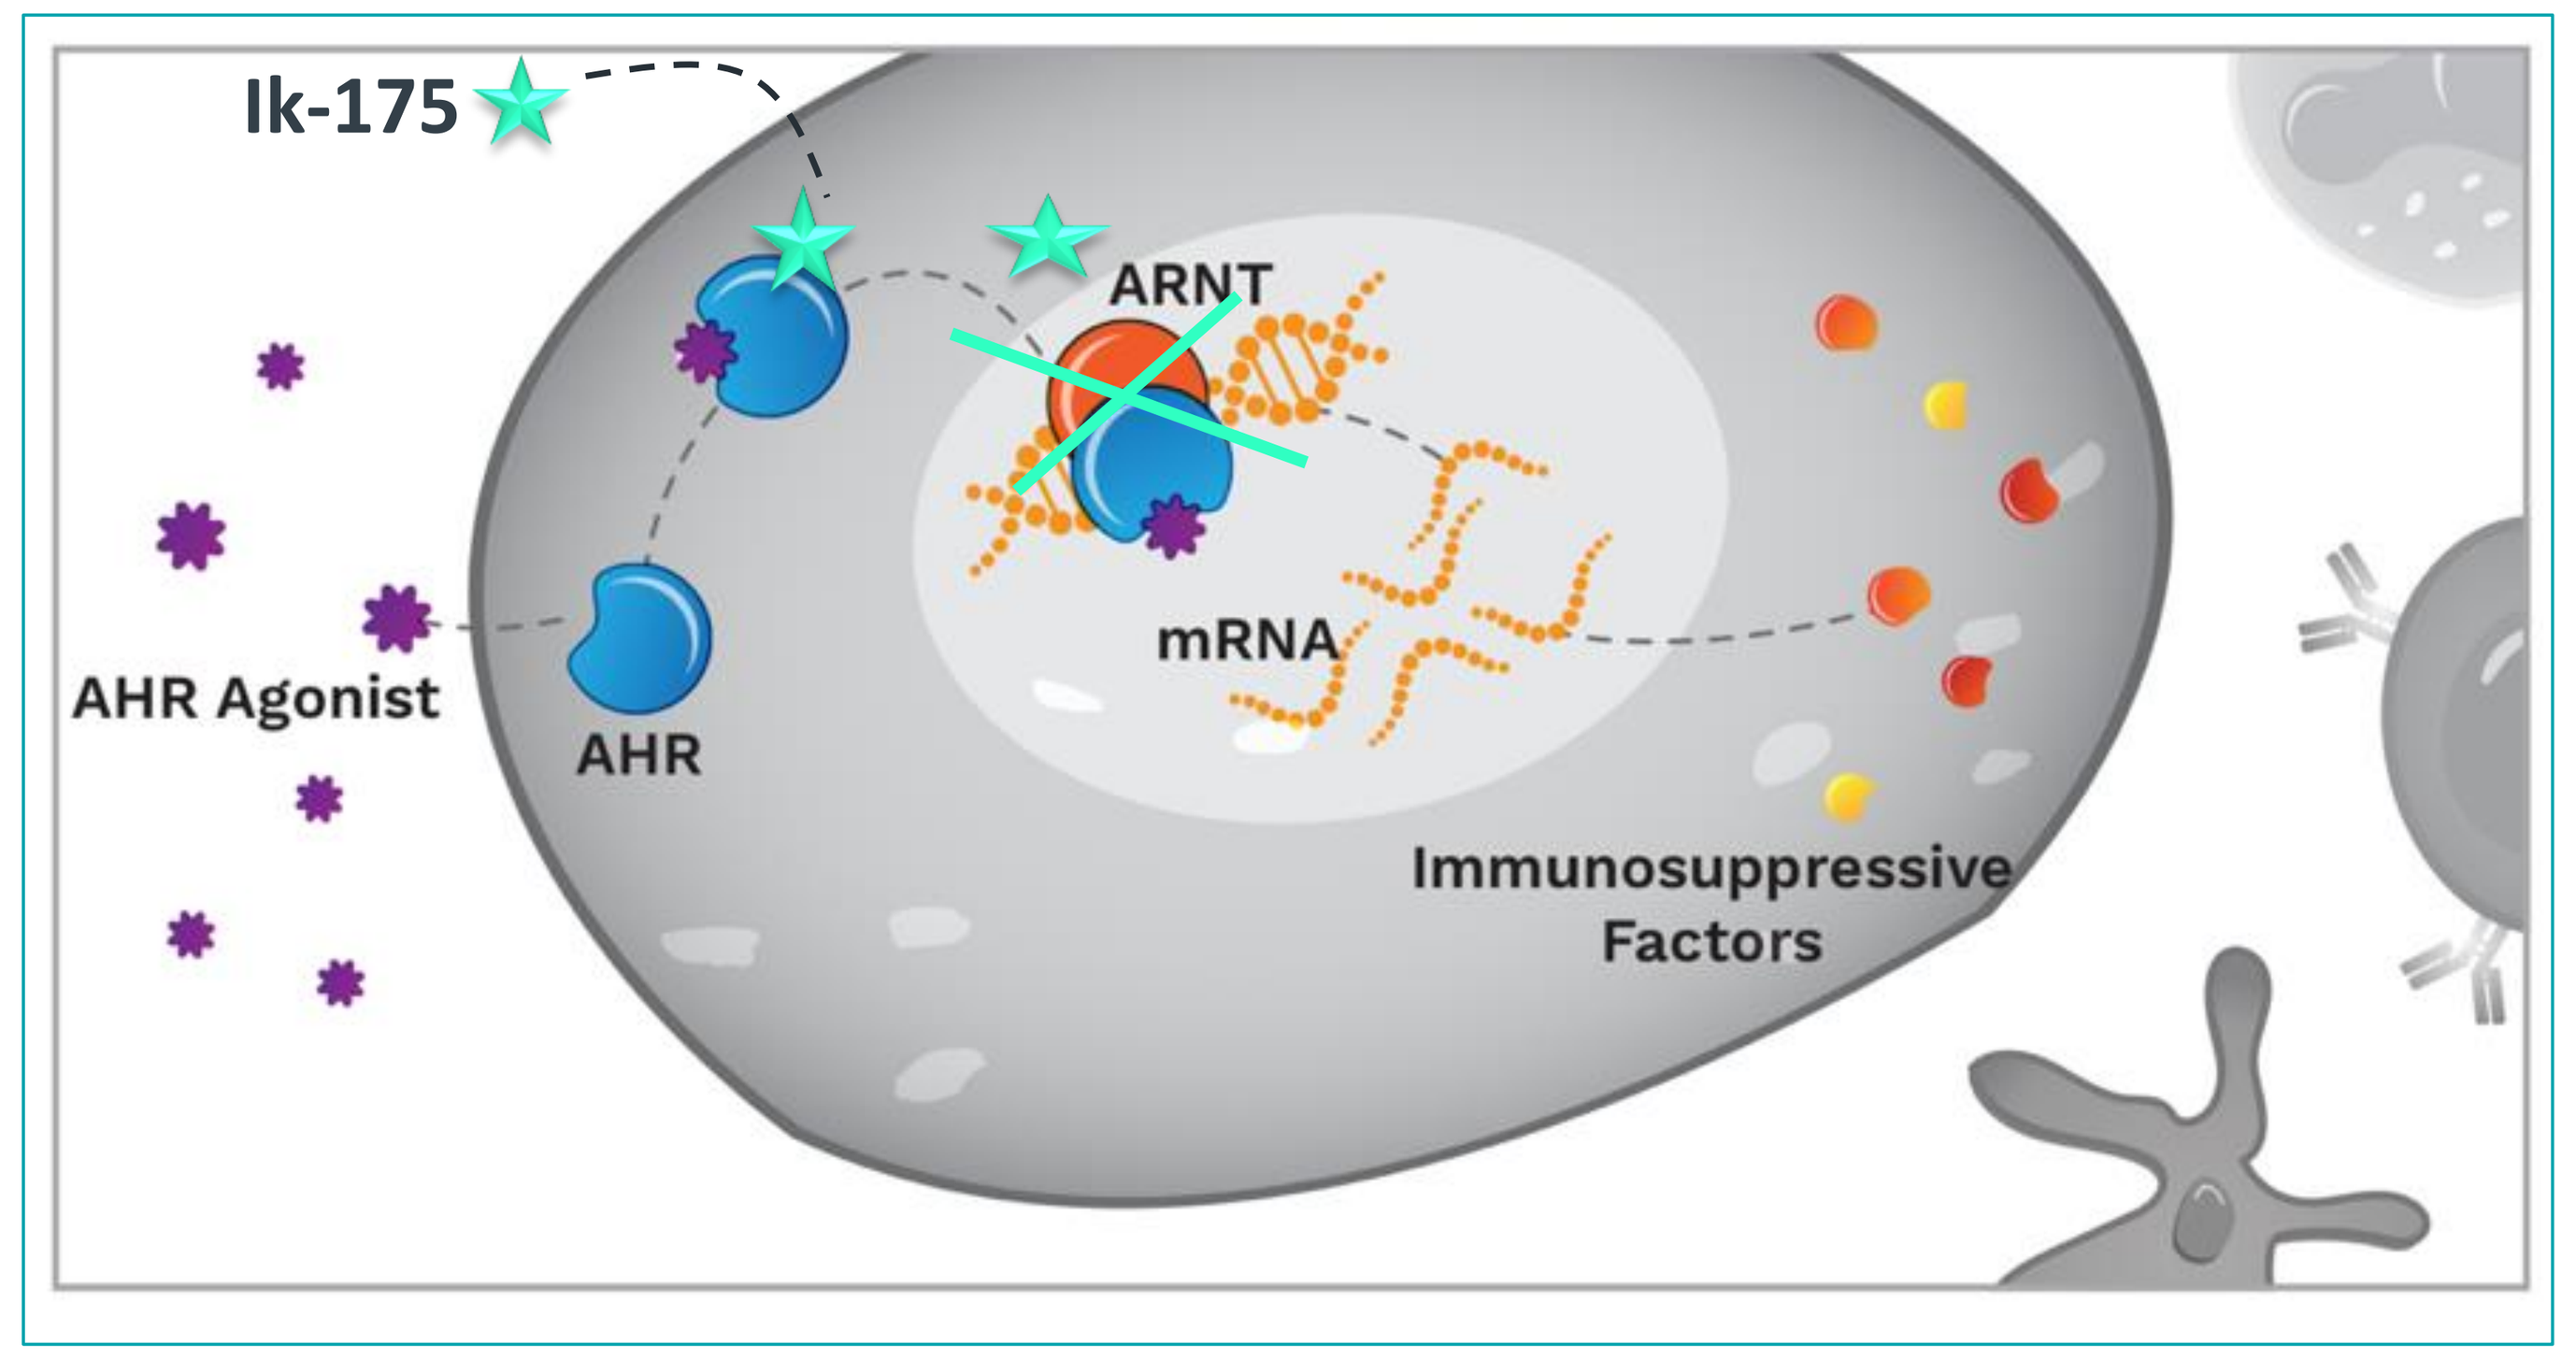
Ik-175
ARNT
mRNA
AHR Agonist
AHR
Immunosuppressive
Factors

AHR activation is implicated in tumor development in multiple cancer types. In addition, high levels of serum kynurenine are associated with resistance to checkpoint inhibitors (Li 2019). To overcome AHR-mediated immunosuppression in cancers, we developed a selective oral AHR inhibitor IK-175 and took a combined computational and tissue-based approach to select cancer indications for its clinical development.

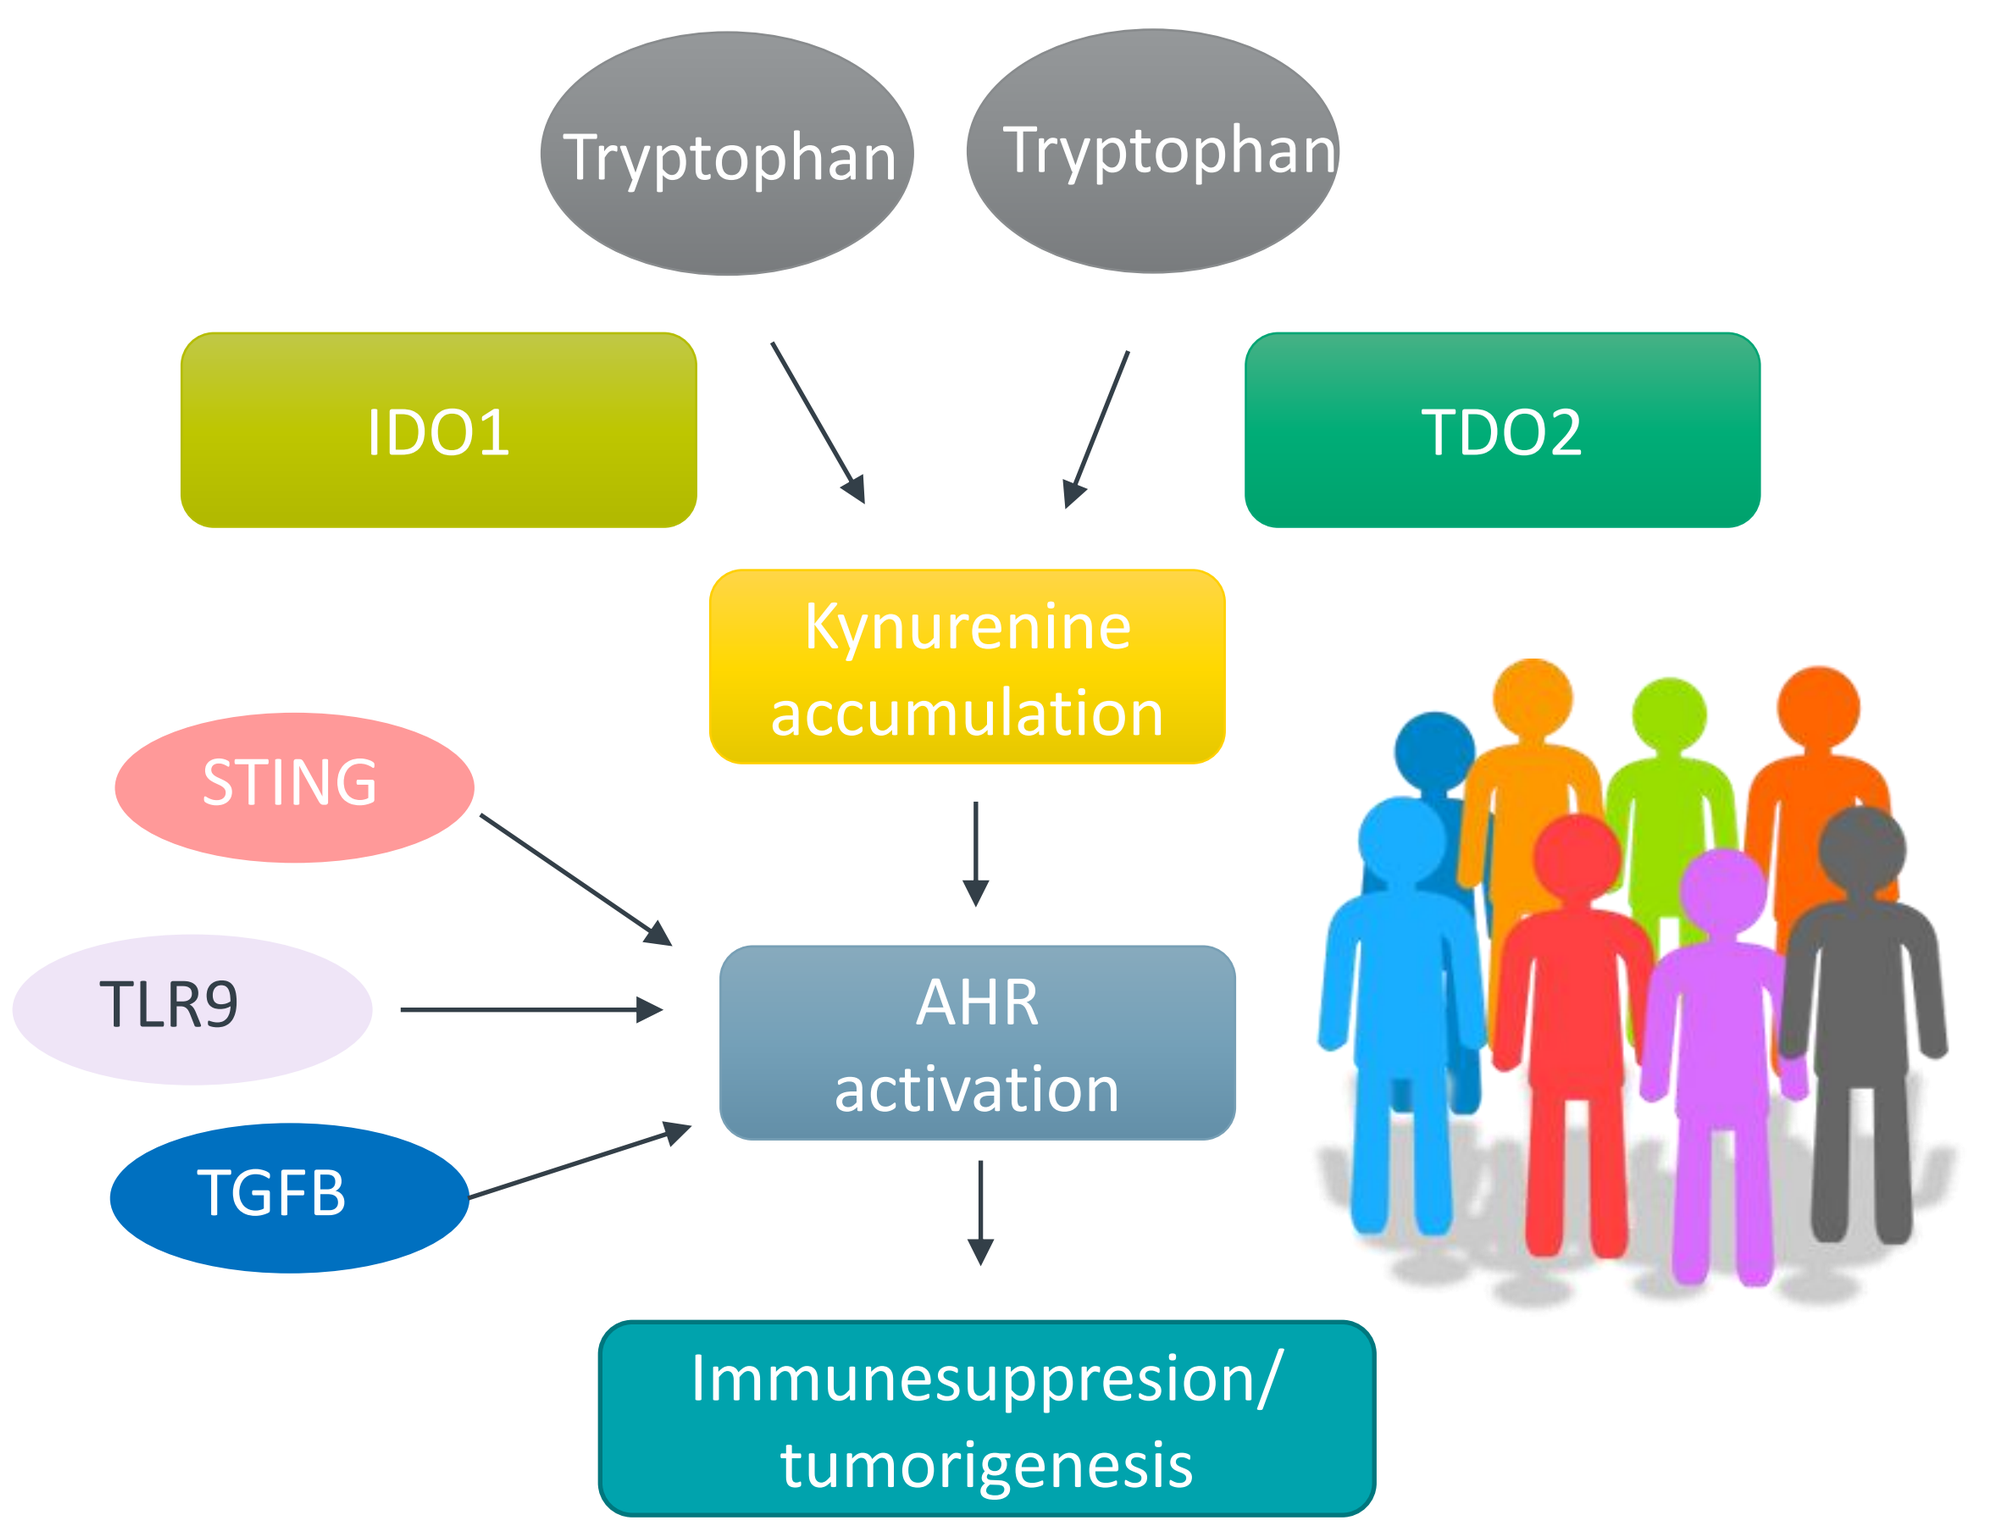
Tryptophan
Tryptophan
IDO1
TDO2
Kynurenine accumulation
STING
TLR9
TGFB
AHR activation
Immunesuppresion/ tumorigenesis

The aim of this work is to identify tumor indications dependent on AHR signaling based on computational and tissue analyses and define patient selection strategies to evaluate AHR inhibition in cancers.

Genomic profiling of solid and hematological cancers from TCGA and Project GENIE demonstrated bladder cancer as the tumor type with the highest prevalence of AHR amplification and the second highest of AHR expression

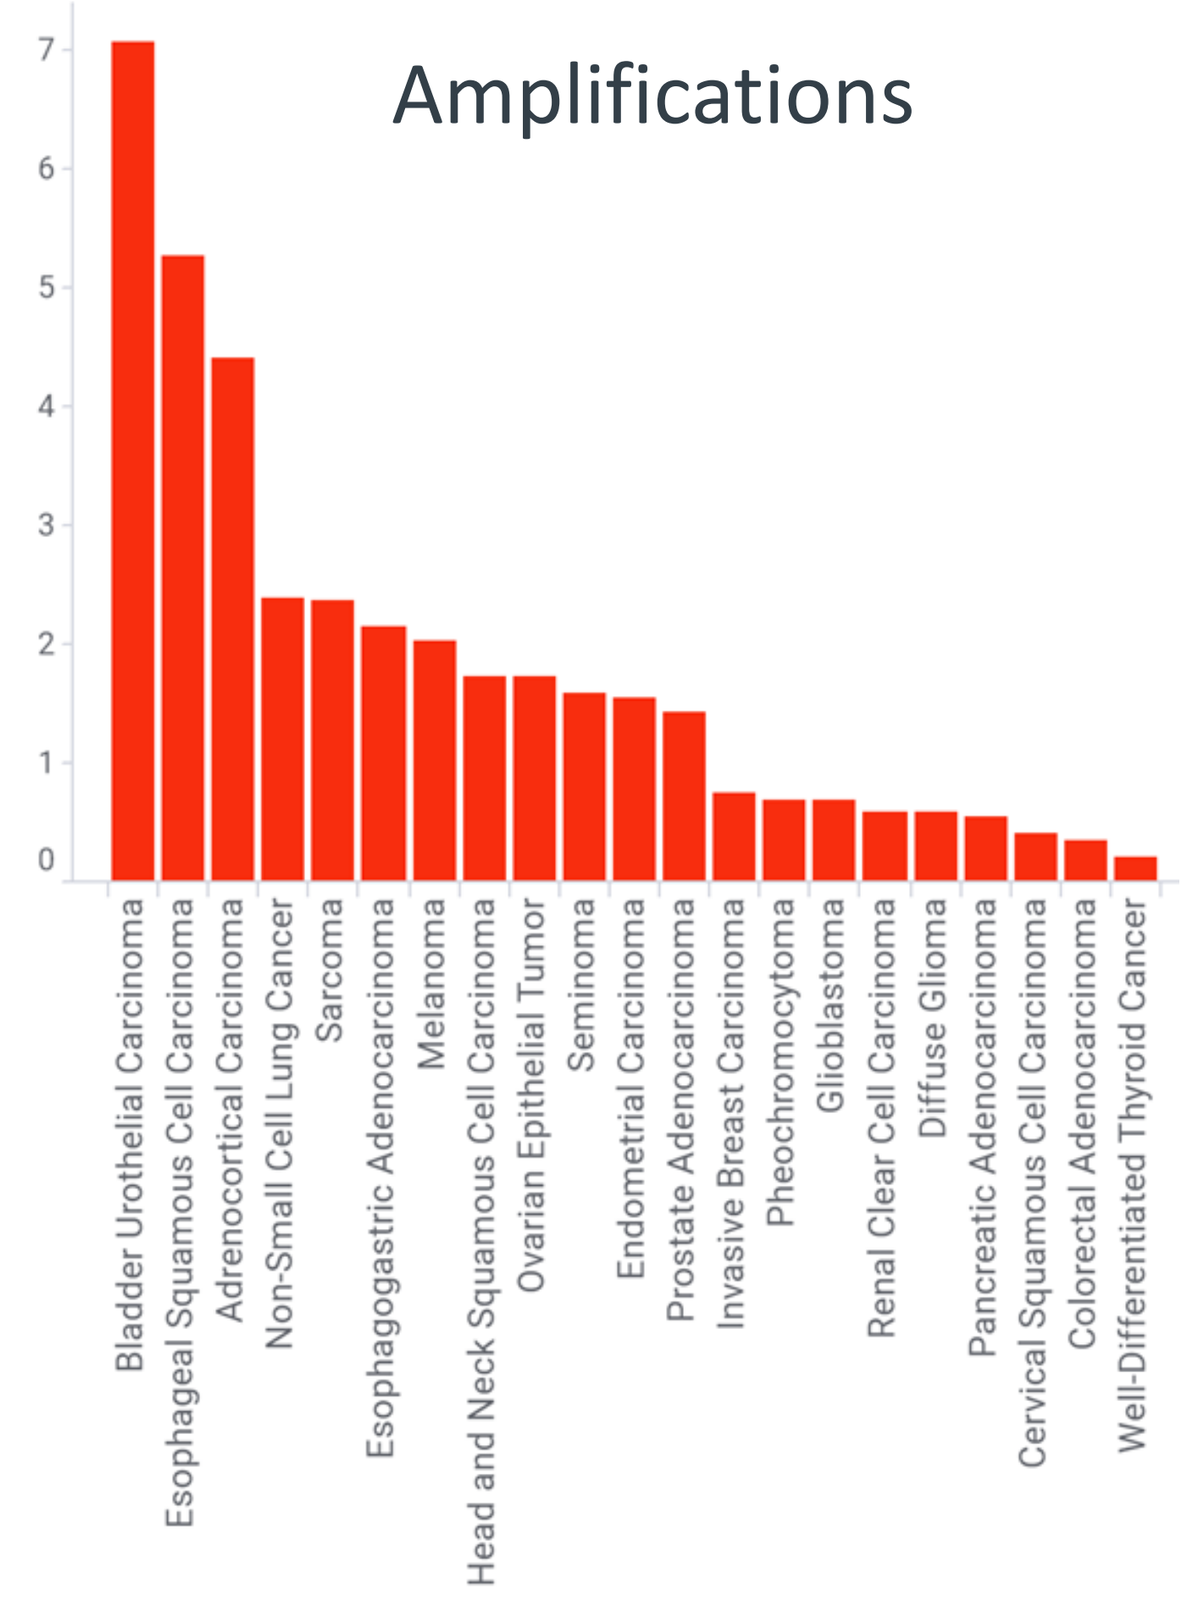
Amplifications
Bladder Urothelial Carcinoma
Esophageal Squamous Cell Carcinoma
Adrenocortical Carcinoma
Non-Small Cell Lung Cancer
Sarcoma
Esophagogastric Adenocarcinoma
Melanoma
Head and Neck Squamous Cell Carcinoma
Ovarian Epithelial Tumor
Seminoma
Endometrial Carcinoma
Prostate Adenocarcinoma
Invasive Breast Carcinoma
Pheochromocytoma
Glioblastoma
Renal Clear Cell Carcinoma
Diffuse Glioma
Pancreatic Adenocarcinoma
Cervical Squamous Cell Carcinoma
Colorectal Adenocarcinoma
Well-Differentiated Thyroid Cancer

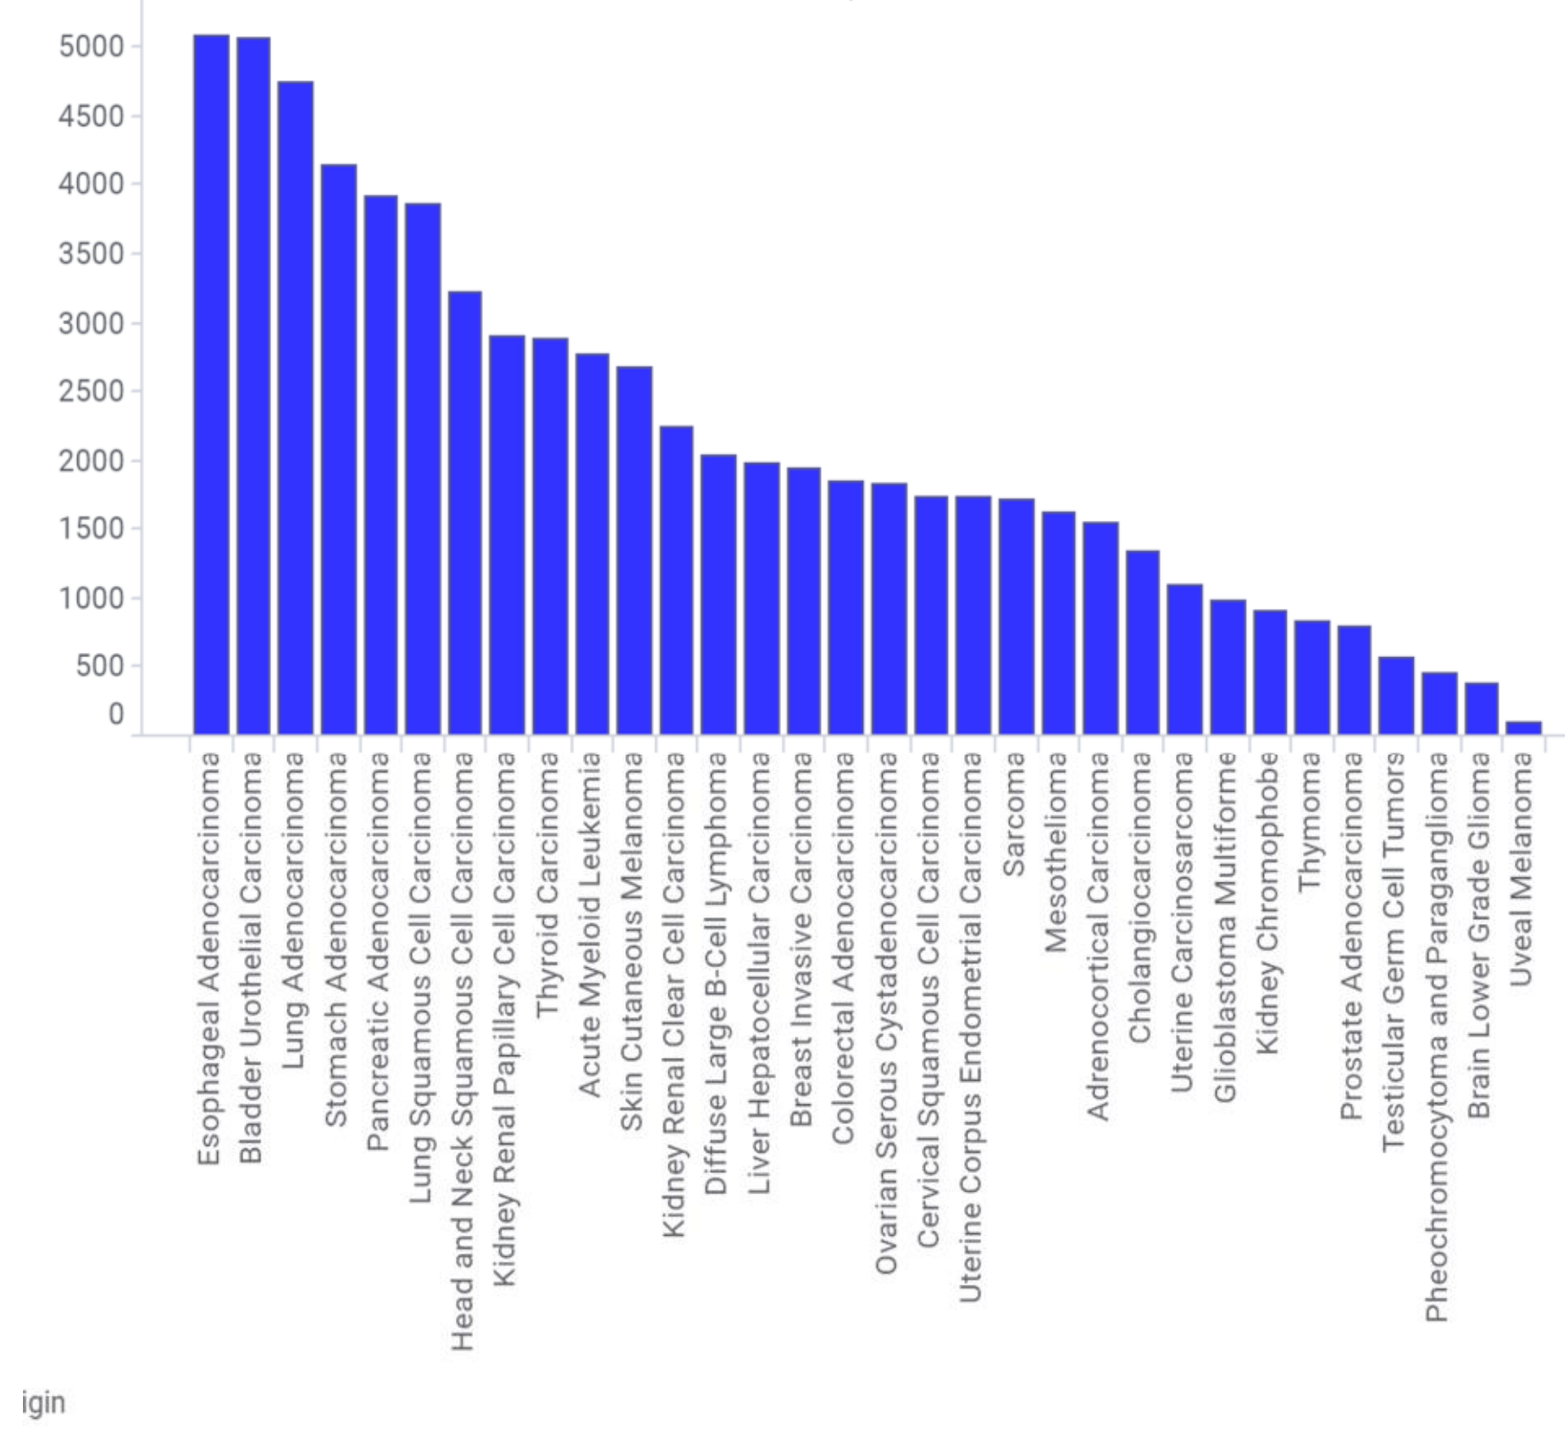
mRNA Expression
Esophageal Adenocarcinoma
Bladder Urothelial Carcinoma
Lung Adenocarcinoma
Stomach Adenocarcinoma
Pancreatic Adenocarcinoma
Lung Squamous Cell Carcinoma
Head and Neck Squamous Cell Carcinoma
Kidney Renal Papillary Cell Carcinoma
Thyroid Carcinoma
Acute Myeloid Leukemia
Skin Cutaneous Melanoma
Kidney Renal Clear Cell Carcinoma
Diffuse Large B-Cell Lymphoma
Liver Hepatocellular Carcinoma
Breast Invasive Carcinoma
Colorectal Adenocarcinoma
Ovarian Serous Cystadenocarcinoma
Cervical Squamous Cell Carcinoma
Uterine Corpus Endometrial Carcinoma
Sarcoma
Mesothelioma
Adrenocortical Carcinoma
Cholangiocarcinoma
Uterine Carcinosarcoma
Glioblastoma Multiforme
Kidney Chromophobe
Thymoma
Prostate Adenocarcinoma
Testicular Germ Cell Tumors
Pheochromocytoma and Paraganglioma
Brain Lower Grade Glioma
Uveal Melanoma

Ranking of tumor types from TCGA and Project GENIE by frequency of copy number gains (left) and mRNA expression level (right) of AHR gene.

AHR signature analysis demonstrated bladder cancer as the tumor type with higher pathway activation

List of AHR interactors by pathway analysis with MetaCORE

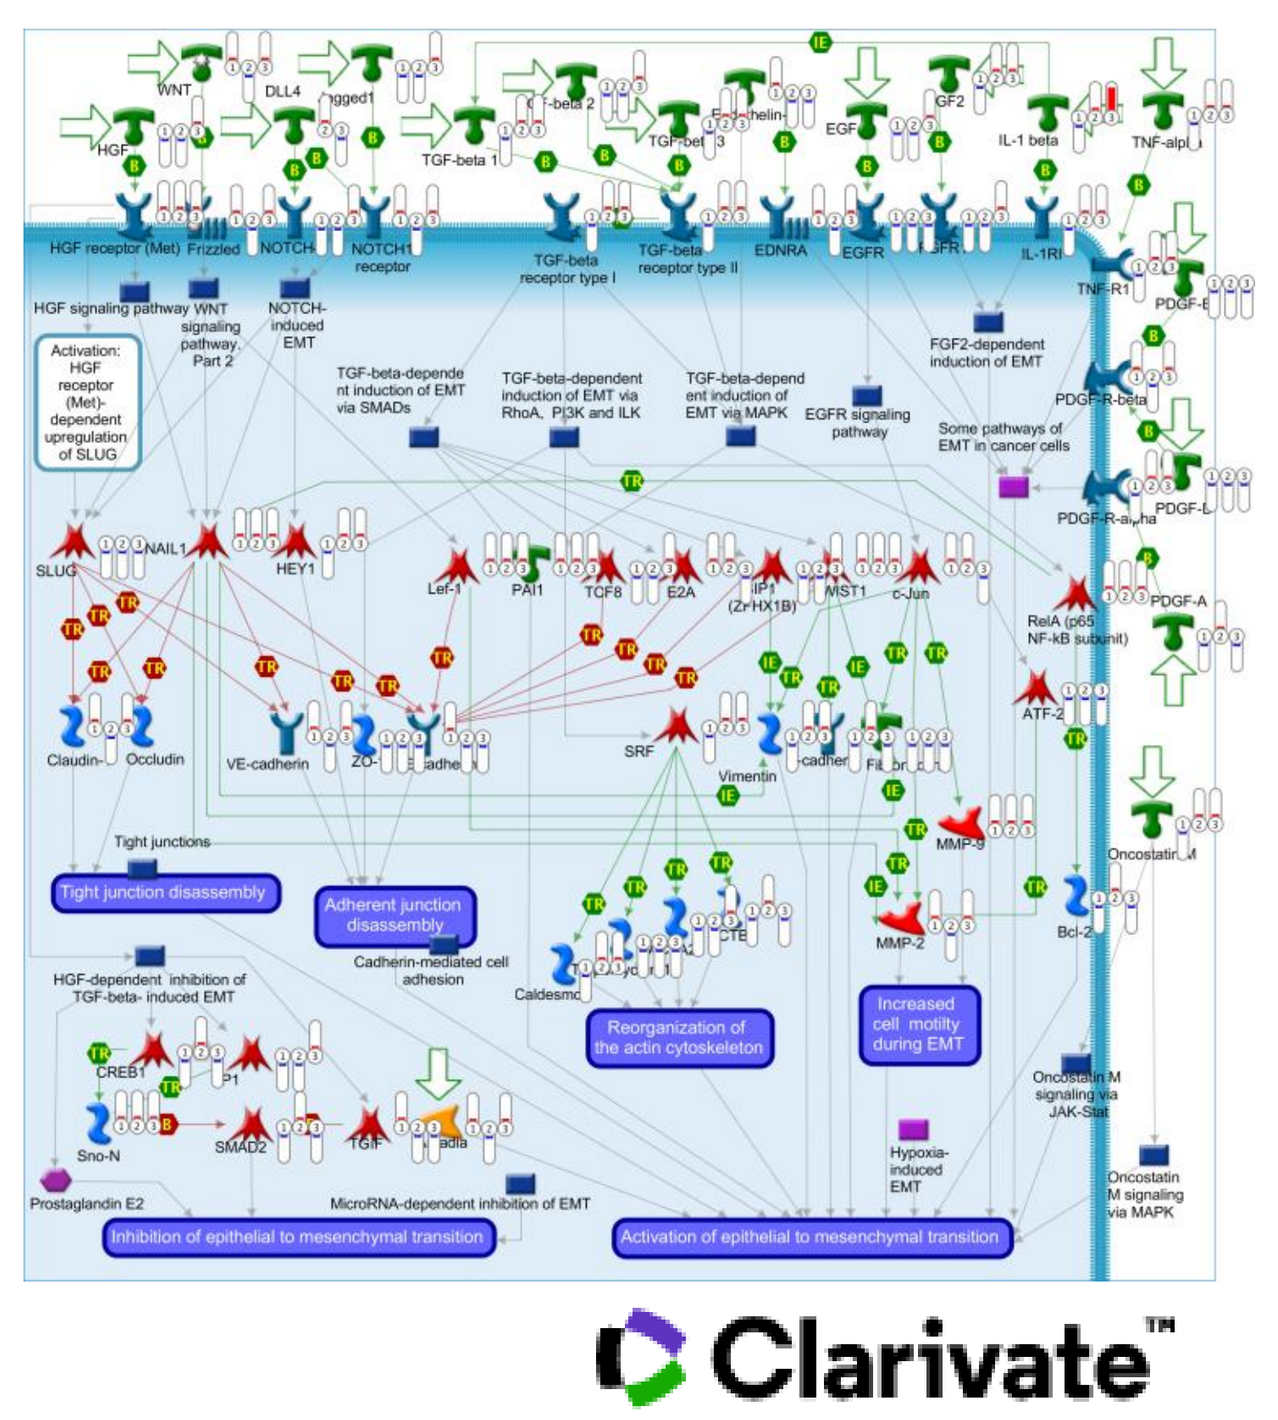
Clarivate™

Modulation of AHR target gene expression across tumor types

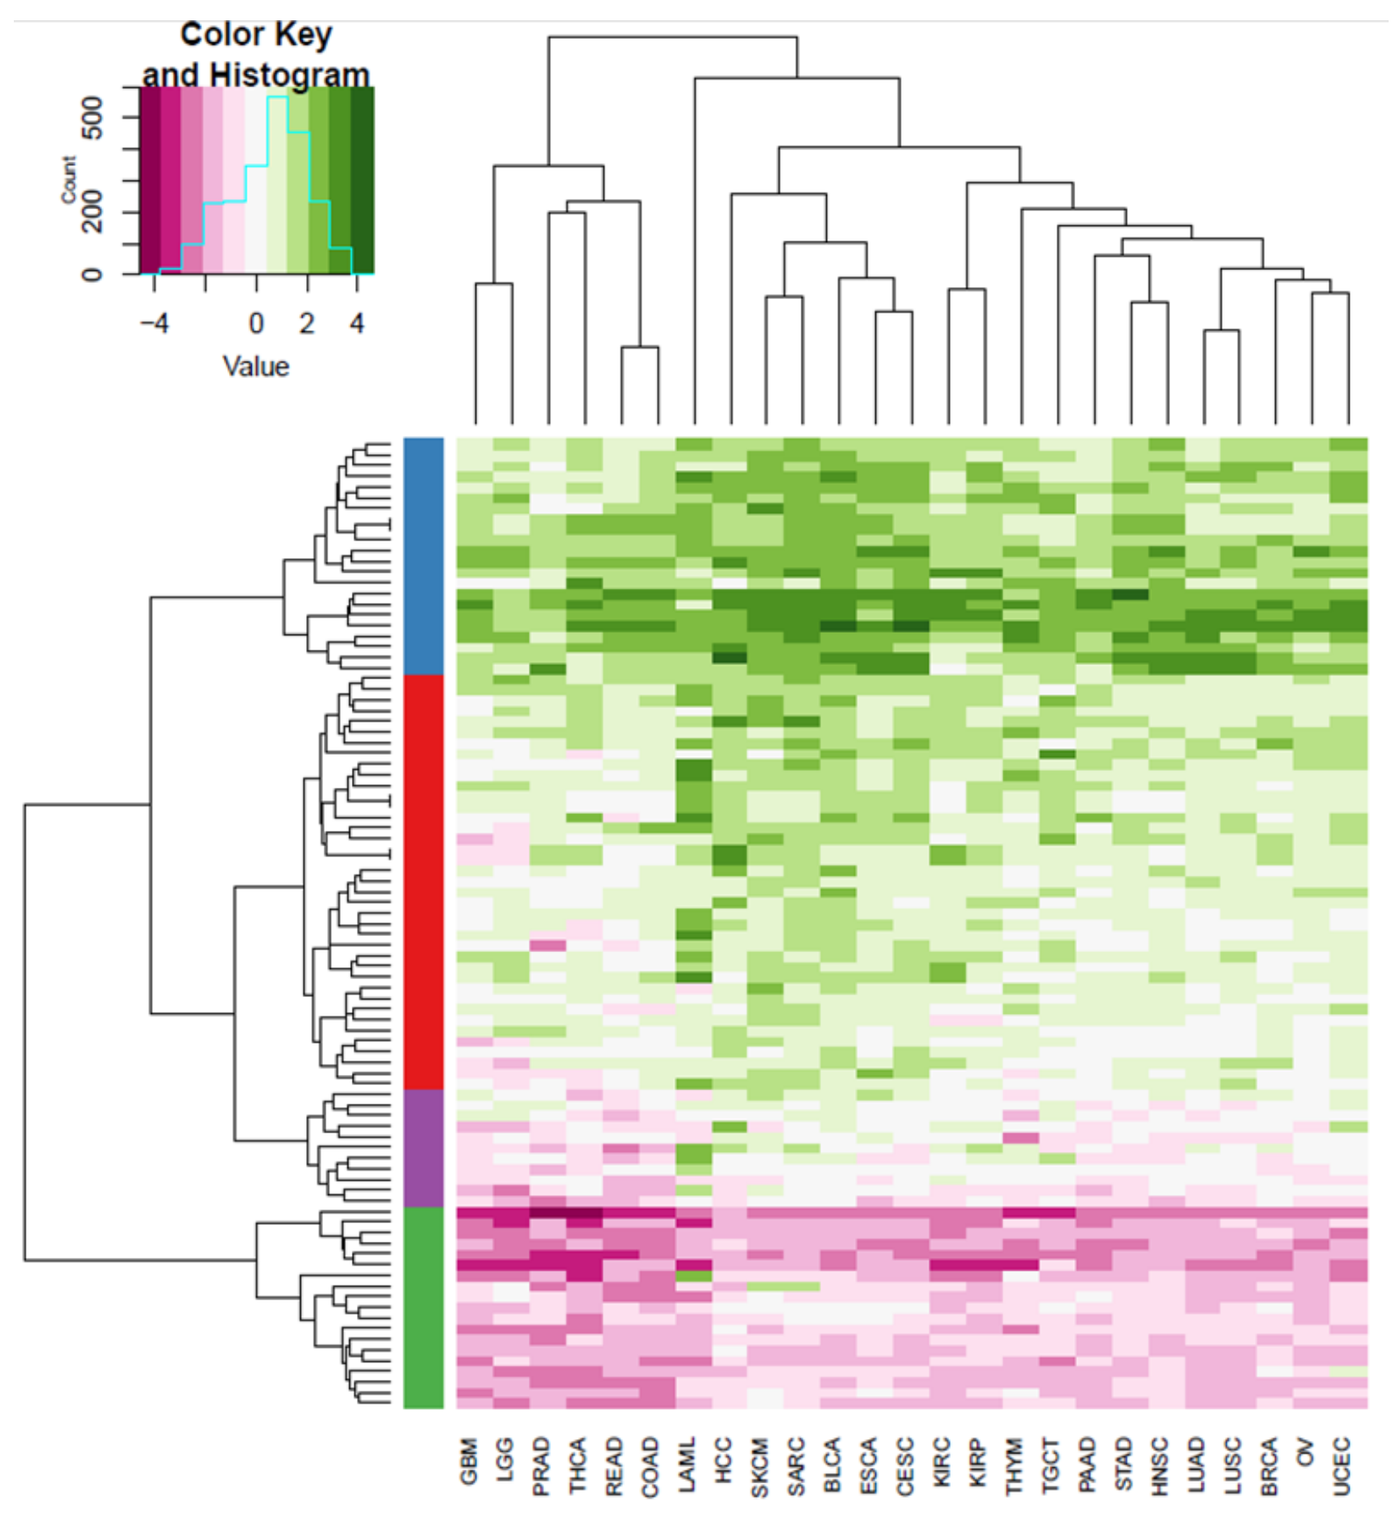
Color Key and Histogram
Count
Value
GBM
LGG
PRAD
THCA
READ
COAD
LAML
HCC
SKCM
SARC
BLCA
ESCA
CESC
KIRC
KIRP
THYM
TGCT
PAAD
STAD
HNSC
LUAD
LUSC
BRCA
OV
UCEC

Tumor types by mean expression of the signature genes

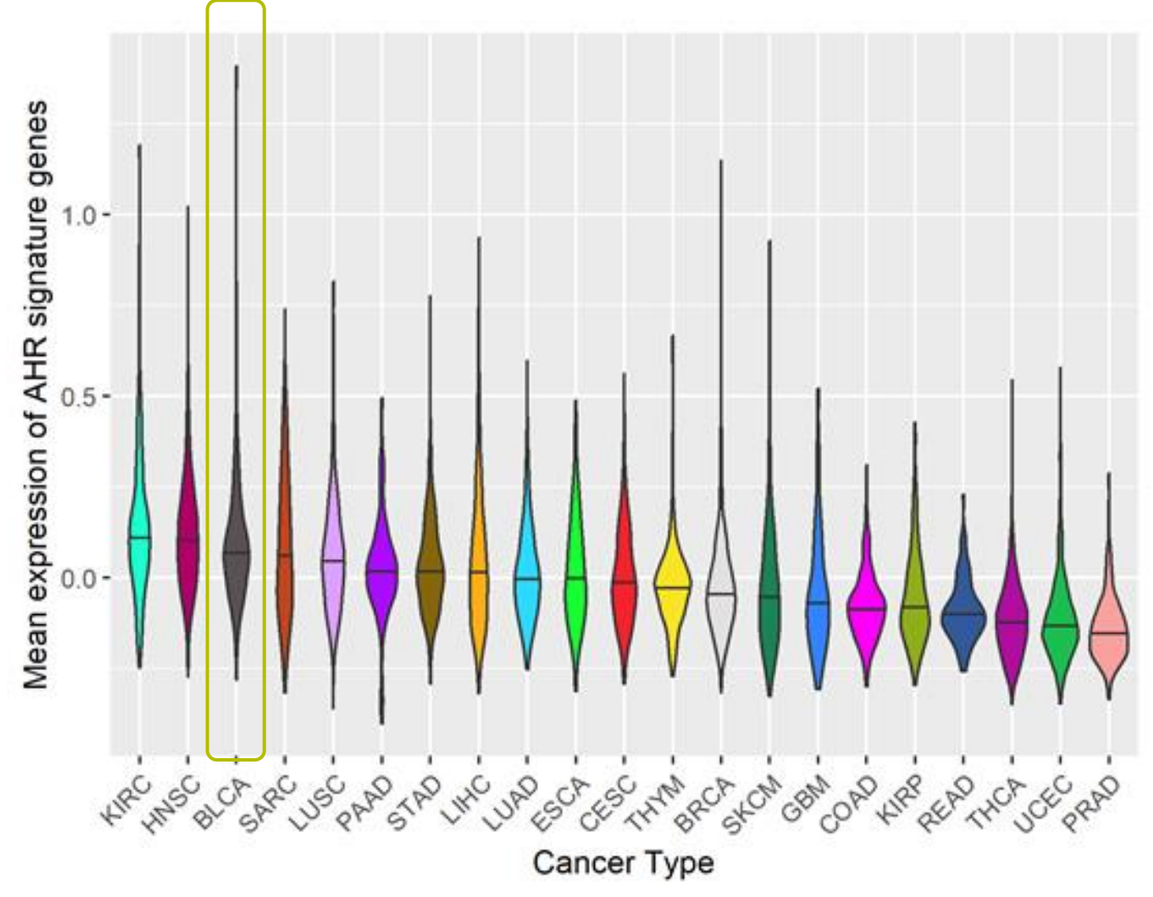
Mean expression of AHR signature genes
Cancer Type
KIRC
HNSC
BLCA
SARC
LUSC
PAAD
STAD
LIHC
LUAD
ESCA
CESC
THYM
BRCA
SKCM
GBM
COAD
KIRP
READ
THCA
UCEC
PRAD

AHR signature correlation with Overall survival in bladder cancer

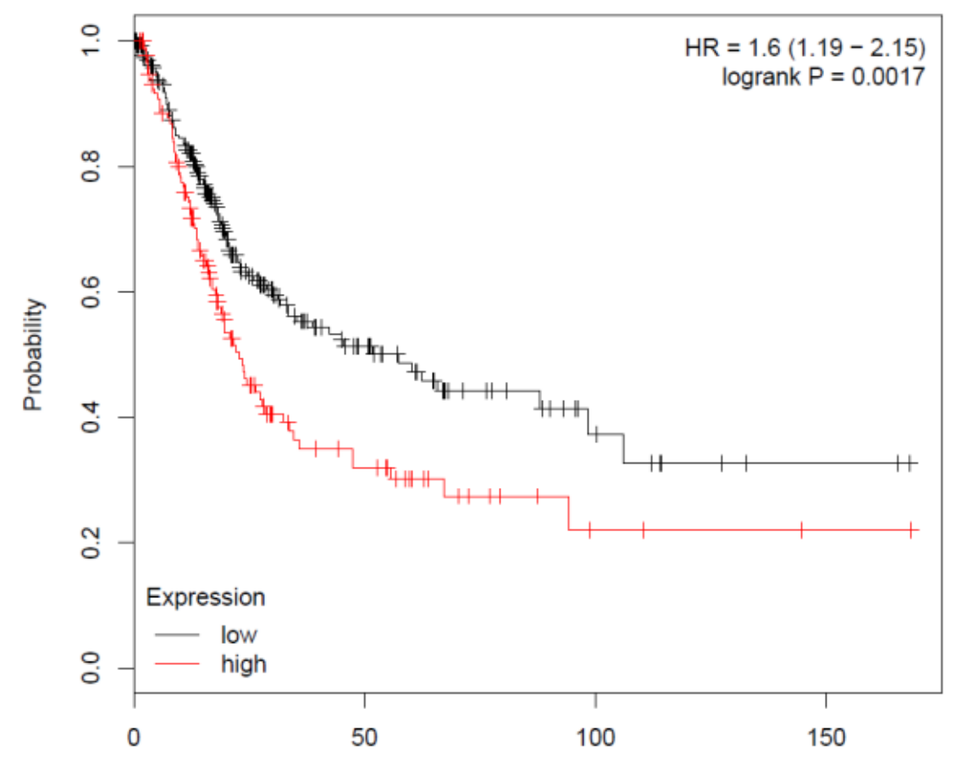
HR = 1.6 (1.19 − 2.15)
logrank P = 0.0017
Probability
Expression
low
high

A proprietary gene signature of AHR activation was developed integrating literature, pathway analysis using MetaCore (Clarivate™) RNAseq and nanostring data from PBMC, T-cells, and cell lines upon AHR inhibition. Transcriptional analysis of the TCGA data using this signature demonstrated high expression of AHR and AHR signature genes in bladder cancer, suggesting increased pathway activity in bladder cancer relative to other cancer types (top 25%). High mean AHR signature gene expression was also associated with worse overall survival in the TCGA bladder cancer cohort (patients were divided by mean expression of signature genes; top 50% vs lowest 50%).

RNAScope analysis of AHR transcripts in tumor and stromal cells from 10 tumor types demonstrated bladder cancer is the tumor type with higher AHR expression

AHR transcripts in Bladder cancer (white)

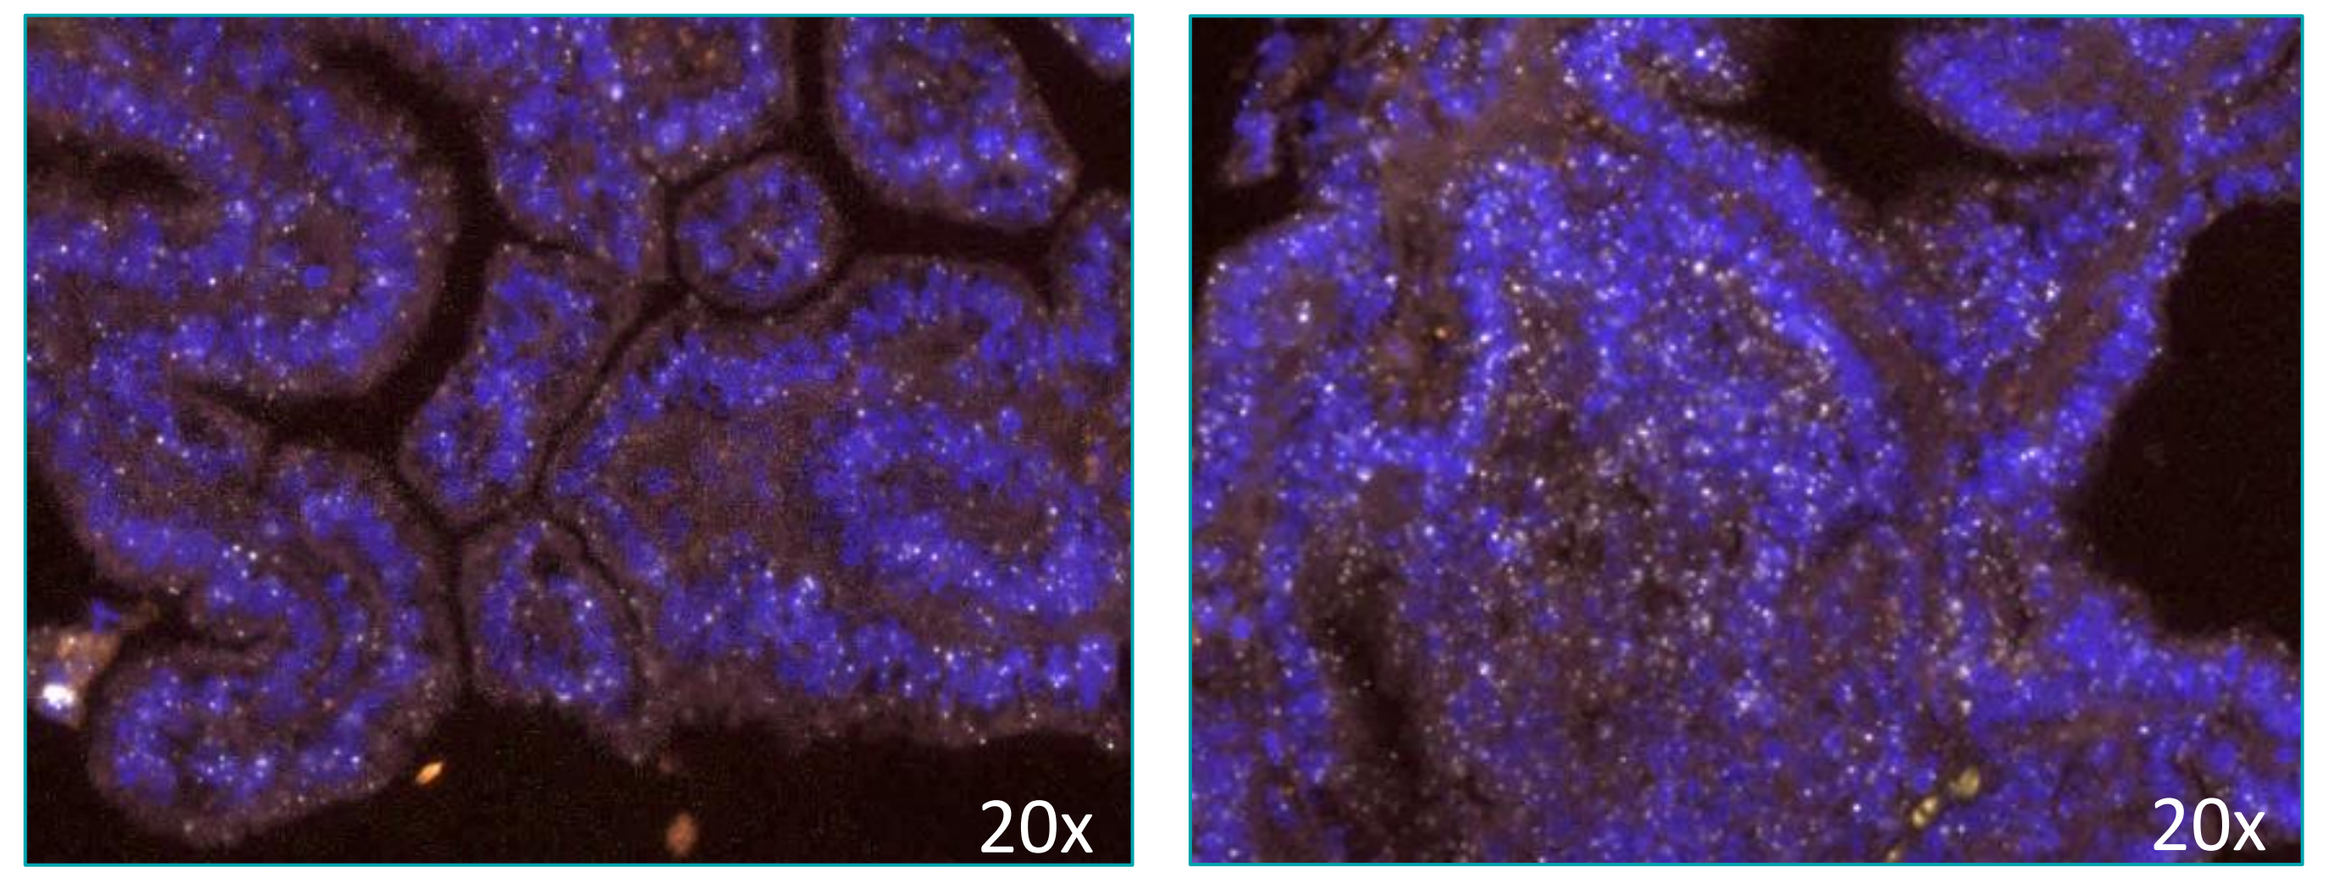
20x
20x

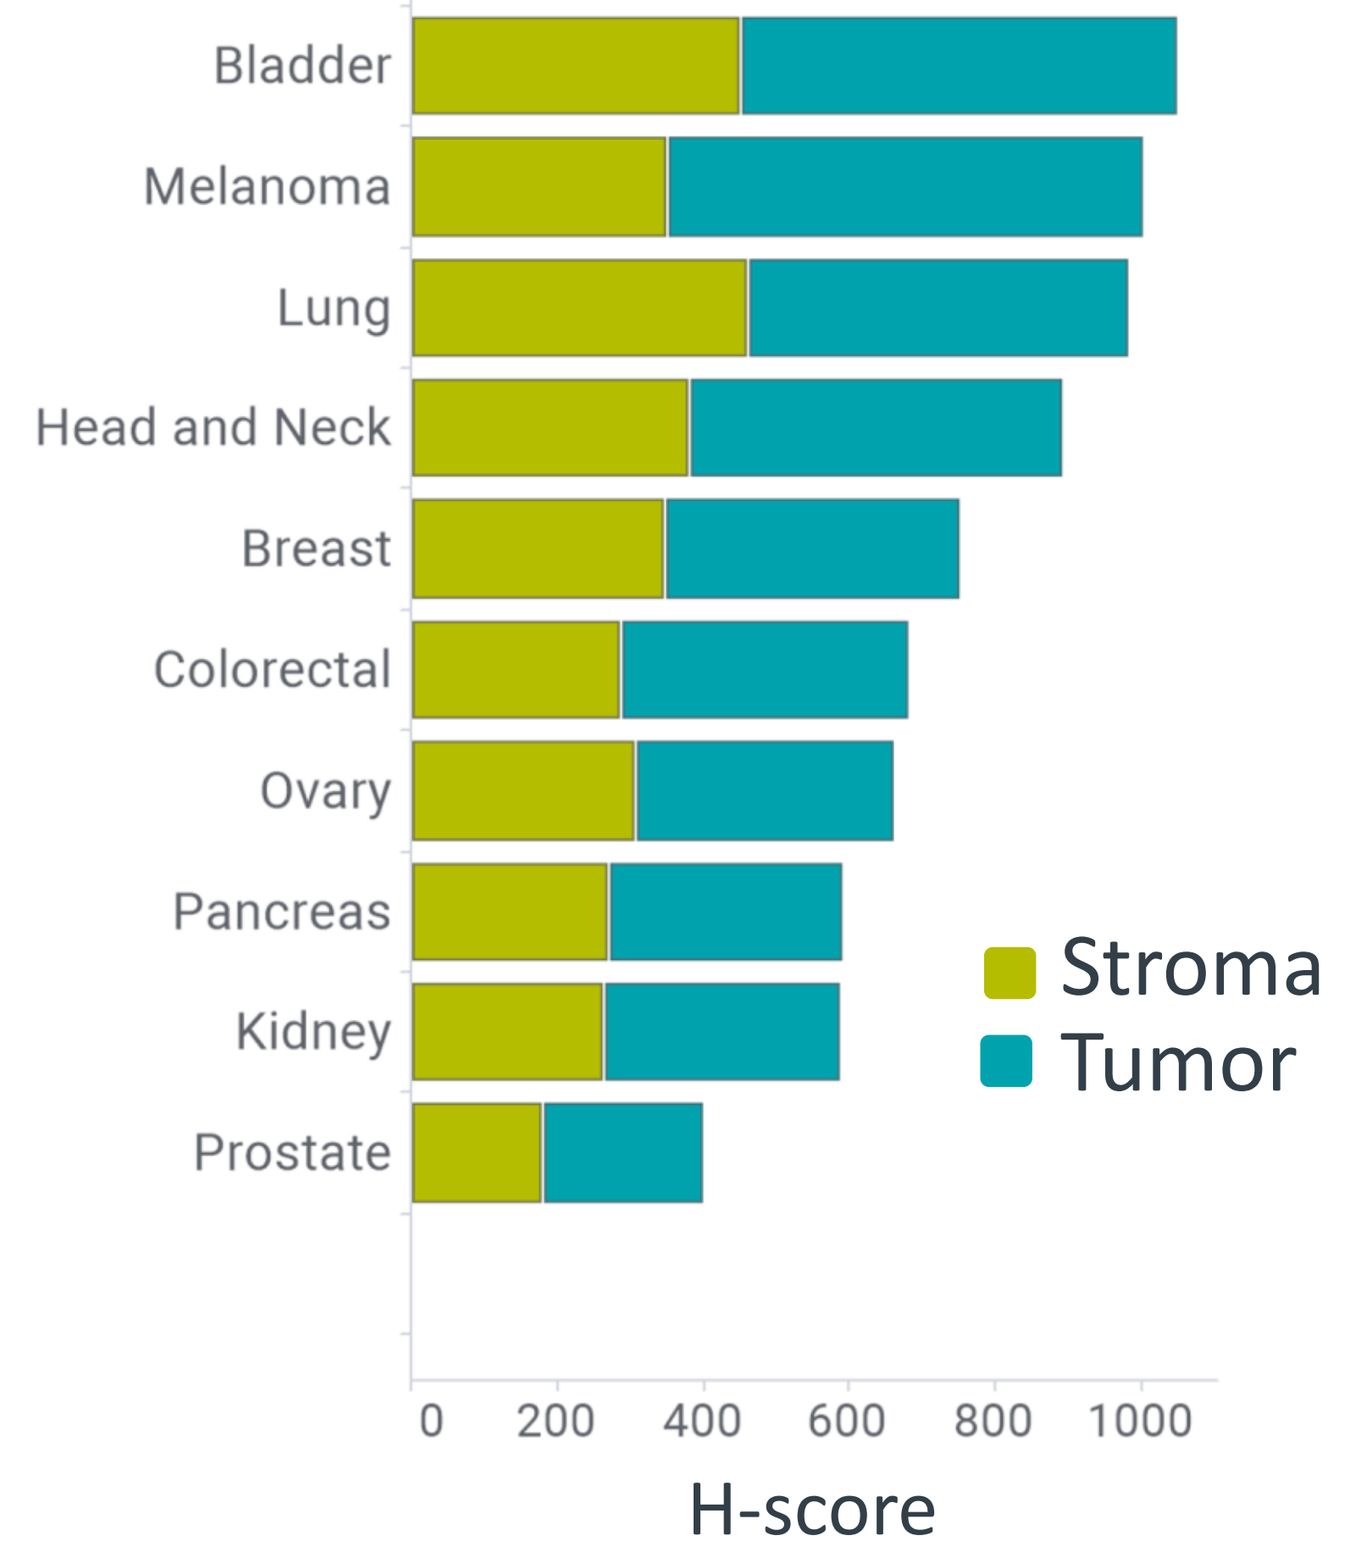
Bladder
Melanoma
Lung
Head and Neck
Breast
Colorectal
Ovary
Pancreas
Kidney
Prostate
Stroma
Tumor
H-score

RNAscope analysis of a tissue microarray containing 10 different tumor types (11 cores per tumor type) revealed bladder cancer has the highest expression of AHR transcripts in the tumor compartment (white dots in the images).

Bladder cancer is the tumor type with highest nuclear AHR protein expression in the tumor region across all cancers evaluated

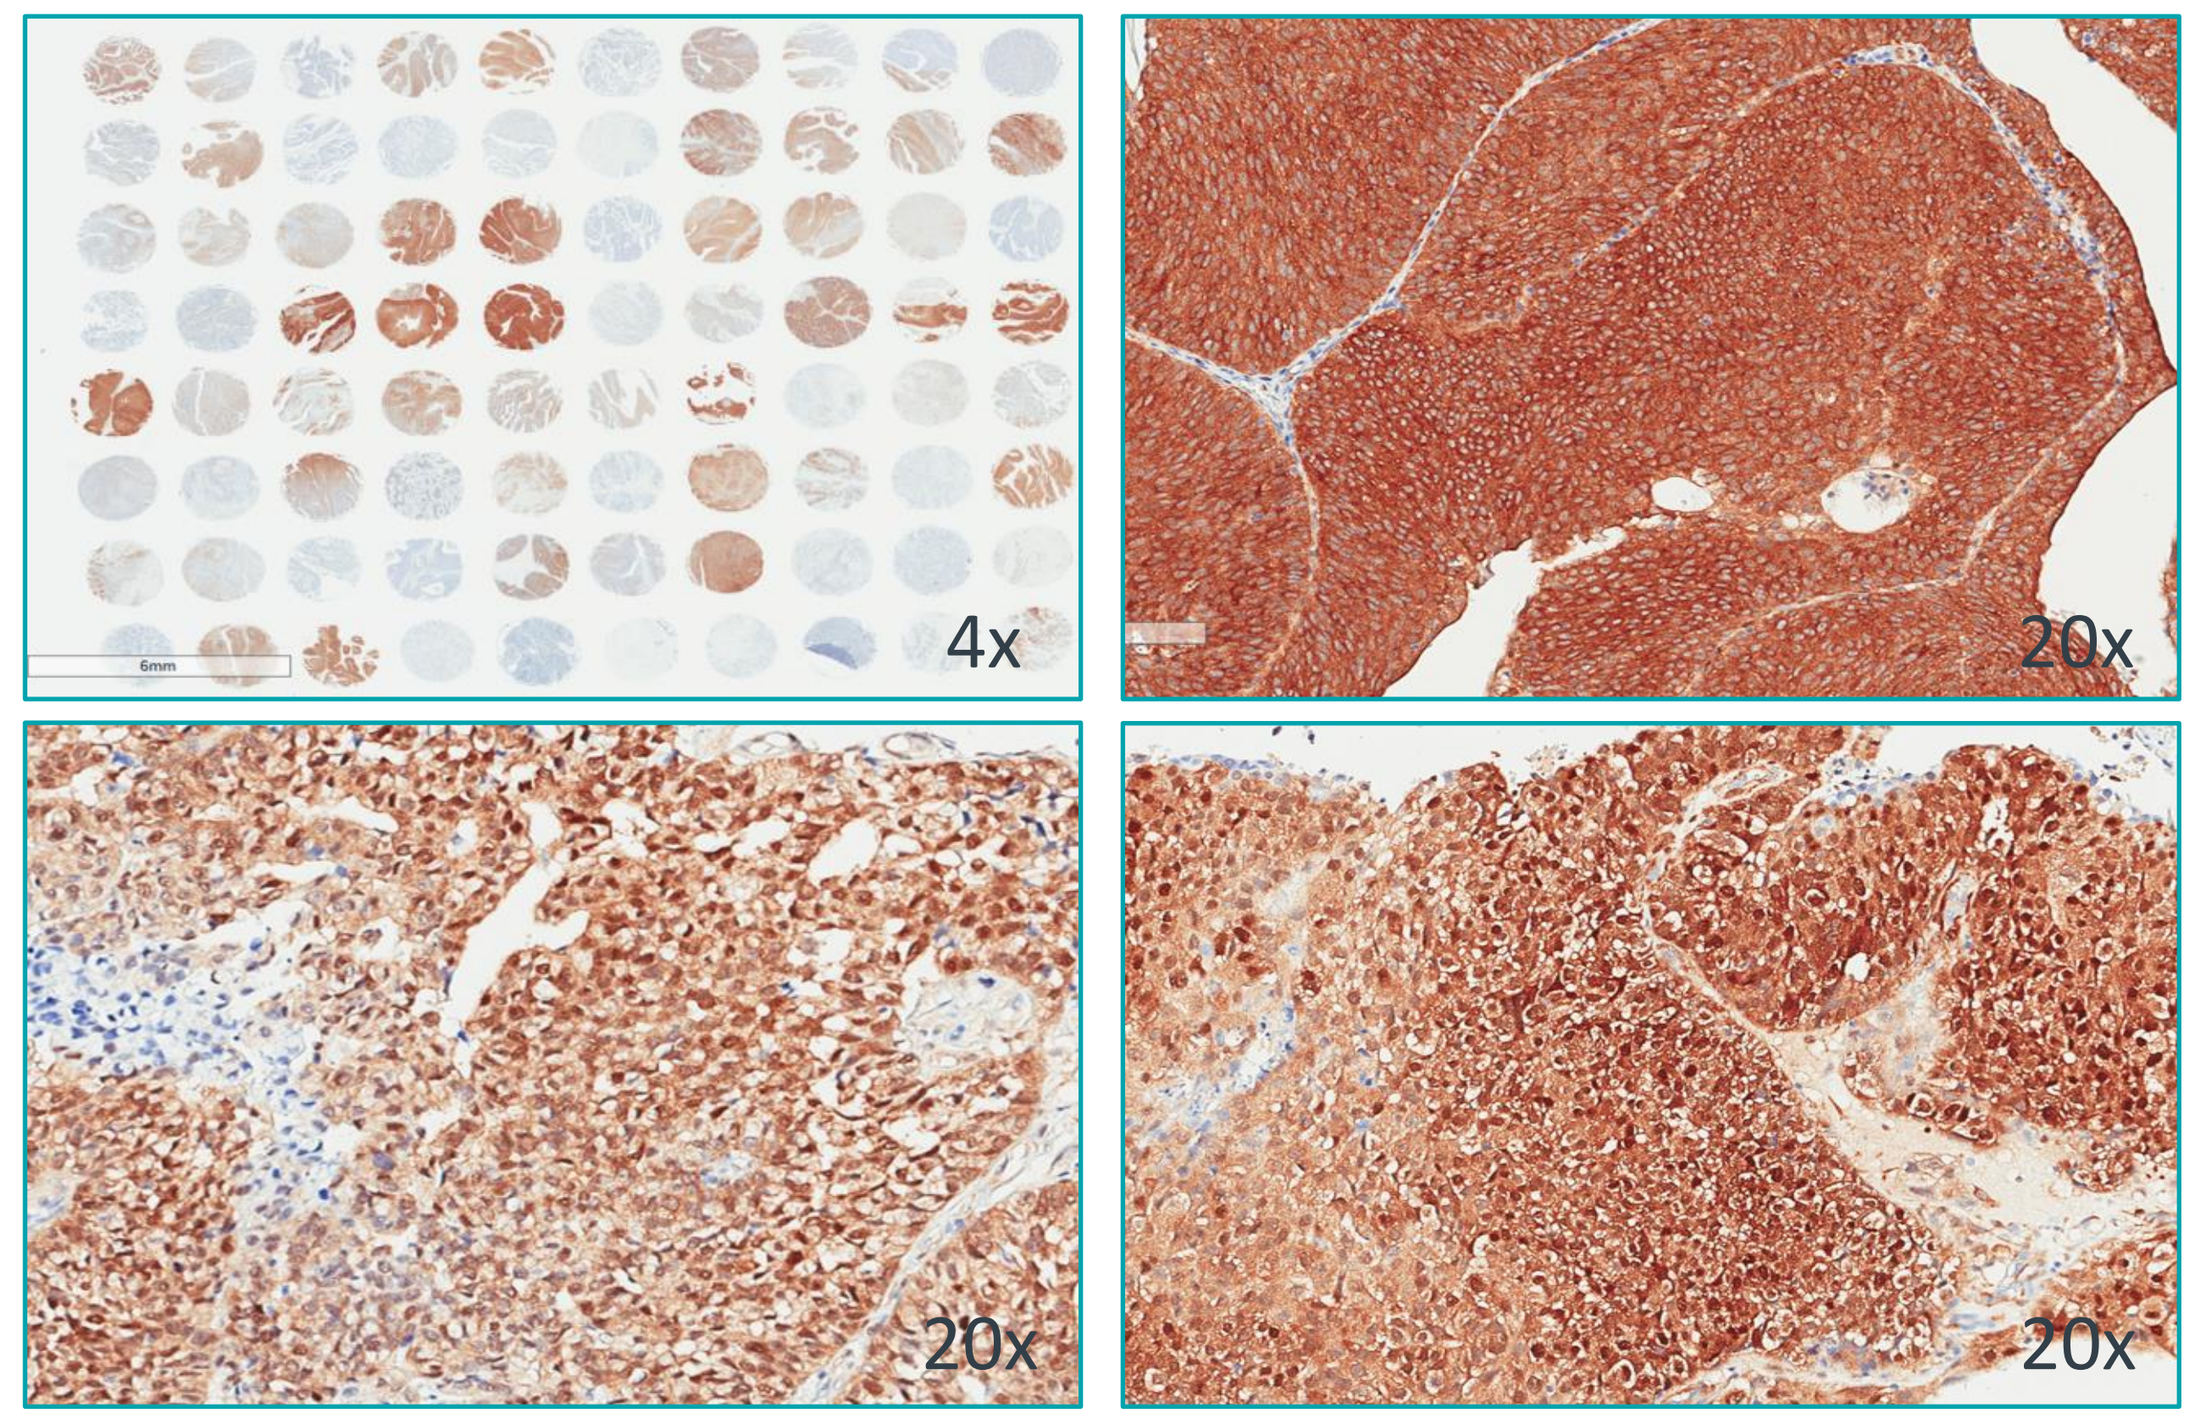
4x
20x
20x
20x

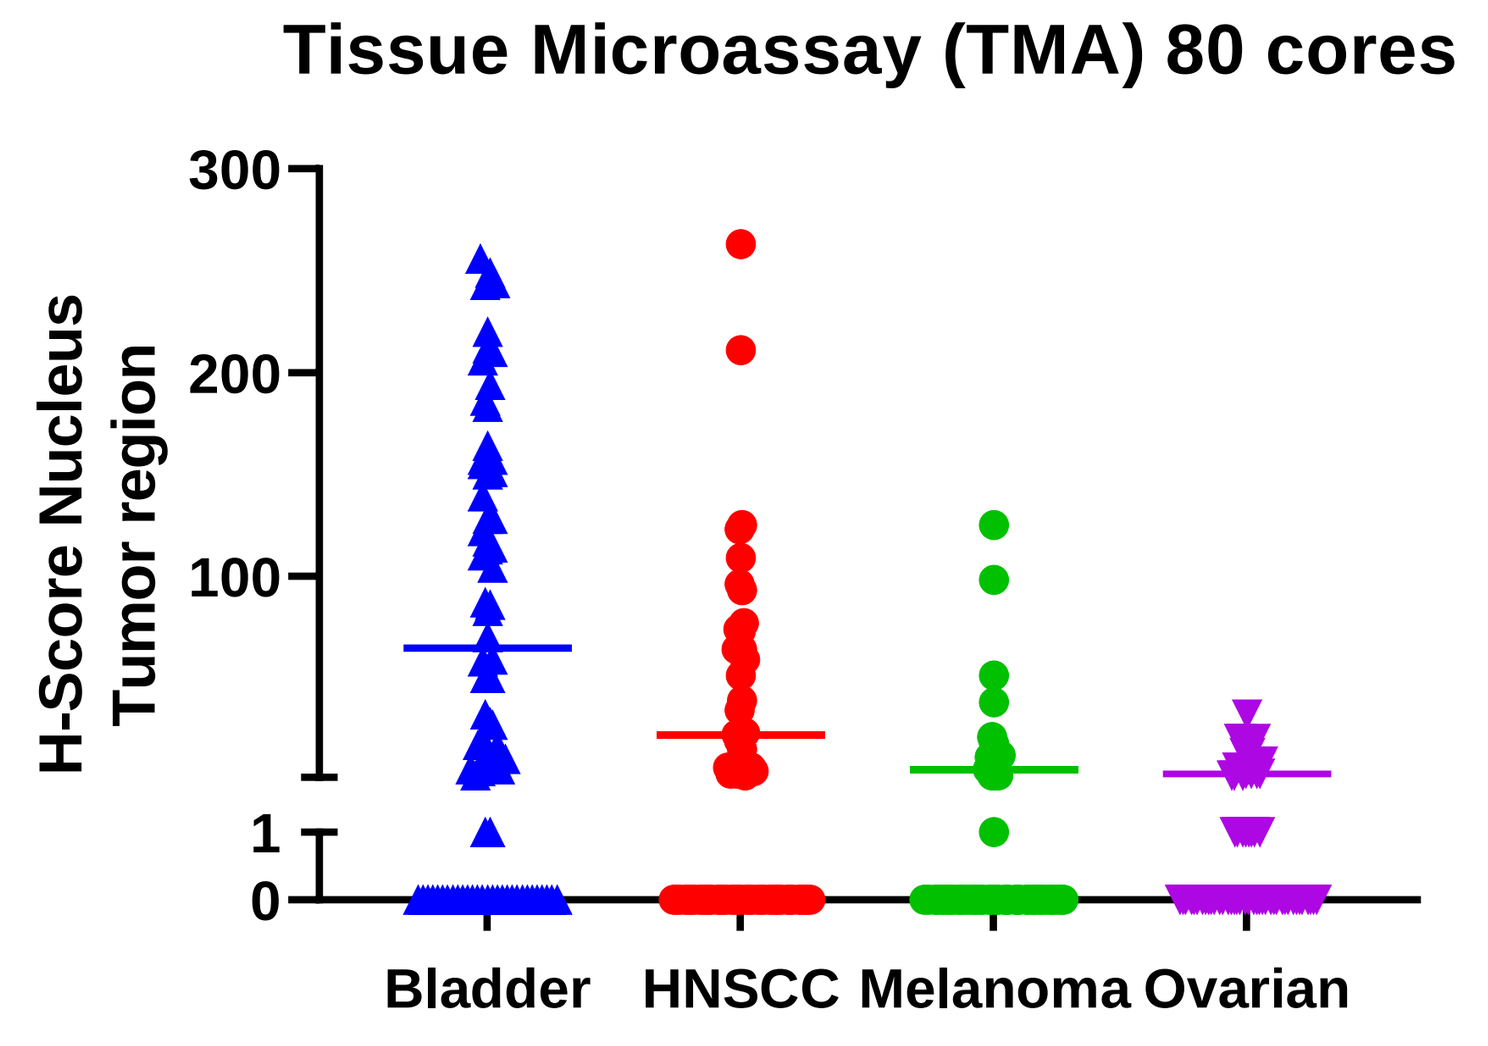
Tissue Microassay (TMA) 80 cores
H-Score Nucleus Tumor region
Bladder
HNSCC
Melanoma
Ovarian

A newly developed IHC assay was used to assess nuclear localization of AHR protein as an indicator of pathway activation. Computational scoring of 4 TMAs from different indications demonstrated bladder cancer as the tumor type with the largest number of high AHR nuclear H-scores.

Conclusions

In summary, we demonstrated high prevalence of nuclear AHR protein expression, AHR gene amplification and target gene expression in bladder cancer, suggesting aberrant AHR activation may play an important role in the progression of this tumor type. This study provides rationale for therapeutic targeting of AHR in bladder cancer patients. Ikena is currently evaluating the anti-tumor activity of IK-175 as a single agent and in combination with nivolumab in bladder cancer in a Phase 1a/1b clinical study (NCT04200963).

correspondence to: msanchez-martin@ikenaoncology.com