#550

TRIAL IN PROGRESS: A Phase 1a/b Study of IK-175, an Oral AHR Inhibitor, Alone and in Combination with Nivolumab in Patients with Locally Advanced or Metastatic Solid Tumors and Urothelial Carcinoma

Meredith A. McKean[1], Jason J. Luke[2], Nehal J. Lakhani[3], Babar Bashir[4], David H. Aggen[5], Alan Tan[6], Katherine Kacena[7], Lei Wang[7], Marissa Timothy[7], and Sergio Santillana[7]

[1]Tennessee Oncology, Nashville, TN, [2]University of Pittsburgh, Pittsburgh, PA, [3]Thomas Jefferson University, Philadelphia, PA, [4]START Midwest, Grand Rapids, MI, [5]Memorial Sloan Kettering Cancer Center, NY, NY, [6]Rush University, Chicago, IL, [7]Ikena Oncology, Boston, MA

ikena ONCOLOGY

BACKGROUND

- Aryl Hydrocarbon Receptor (AHR) is a transcription factor that regulates the activity of multiple innate and adaptive immune cells after binding to several endogenous and exogenous ligands, including kynurenine, generated from the precursor tryptophan by IDO1 and TDO2.
- Binding of kynurenine to AHR leads to a net immunosuppressive tumor microenvironment, making AHR an attractive therapeutic target in multiple cancer types.
- IK-175 was selected based on mechanism of action and robust antitumor activity in preclinical models.
- The combination of IK-175 and nivolumab was selected based on complementary mechanisms of action and robust antitumor activity in preclinical models.
- AHR immunohistochemistry (IHC) tumor microarray analysis across 15 different tumor types revealed that bladder cancer has the highest level of AHR protein expression and AHR nuclear localization, an indicator of active AHR signaling.
- Therefore, nuclear AHR in urothelial carcinoma tumors is being investigated for potential predictive clinical benefit with IK-175.

IK-175 is a selective, small molecule AHR inhibitor being developed as an oral (PO) agent. In human T-cells, IK-175 induces an activated T-cell state, interleukin (IL)-22 gene expression, and leads to an increase in proinflammatory cytokines, such as IL-2 and IL-9. IK-175 demonstrates antitumor activity as a single agent or in combination with anti-PD1 inhibitors in multiple mouse tumor models.

Identification and Characterization of IK -175

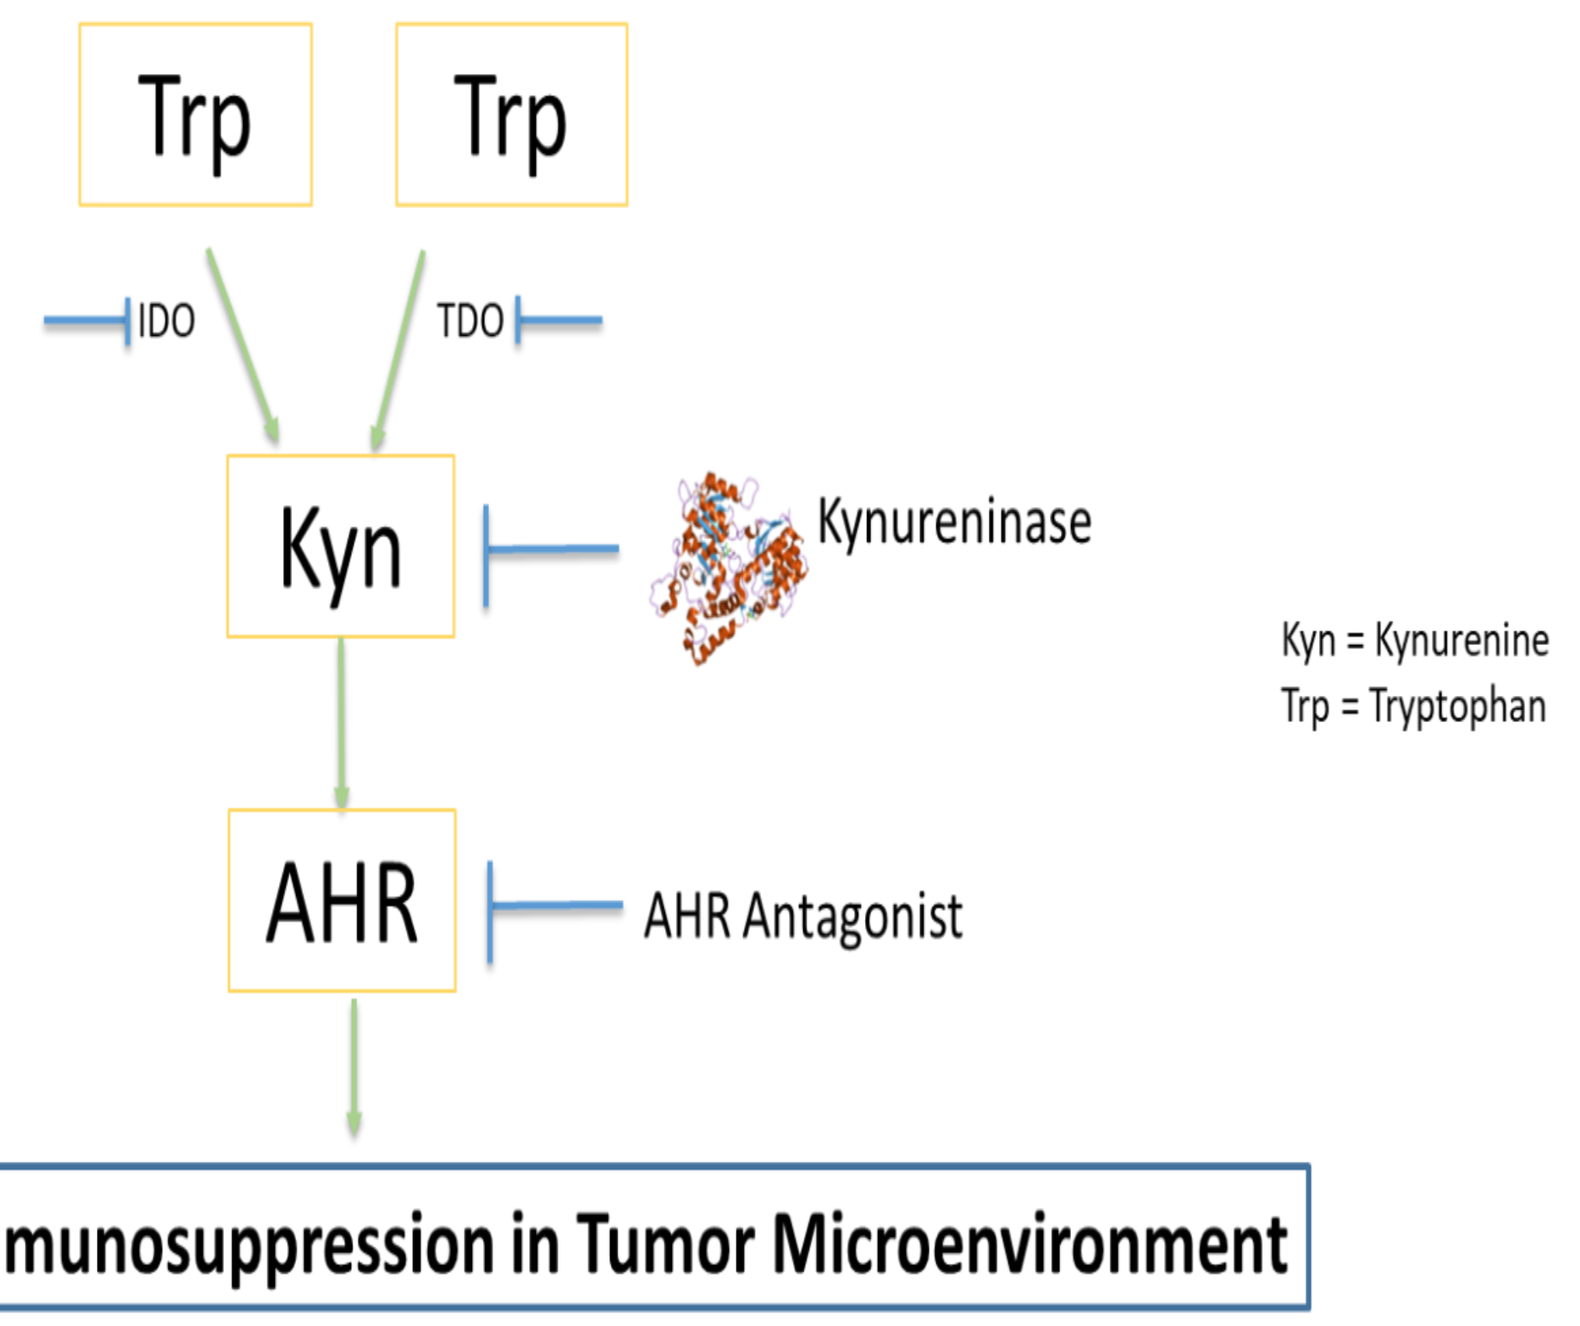
Trp
Trp
IDO
TDO
Kyn
Kynureninase
Kyn = Kynurenine
Trp = Tryptophan
AHR
AHR Antagonist
Broad Immunosuppression in Tumor Microenvironment

IK-175 Single Agent Antitumor Activity and in Combination with anti-PD-1 in a Murine Model

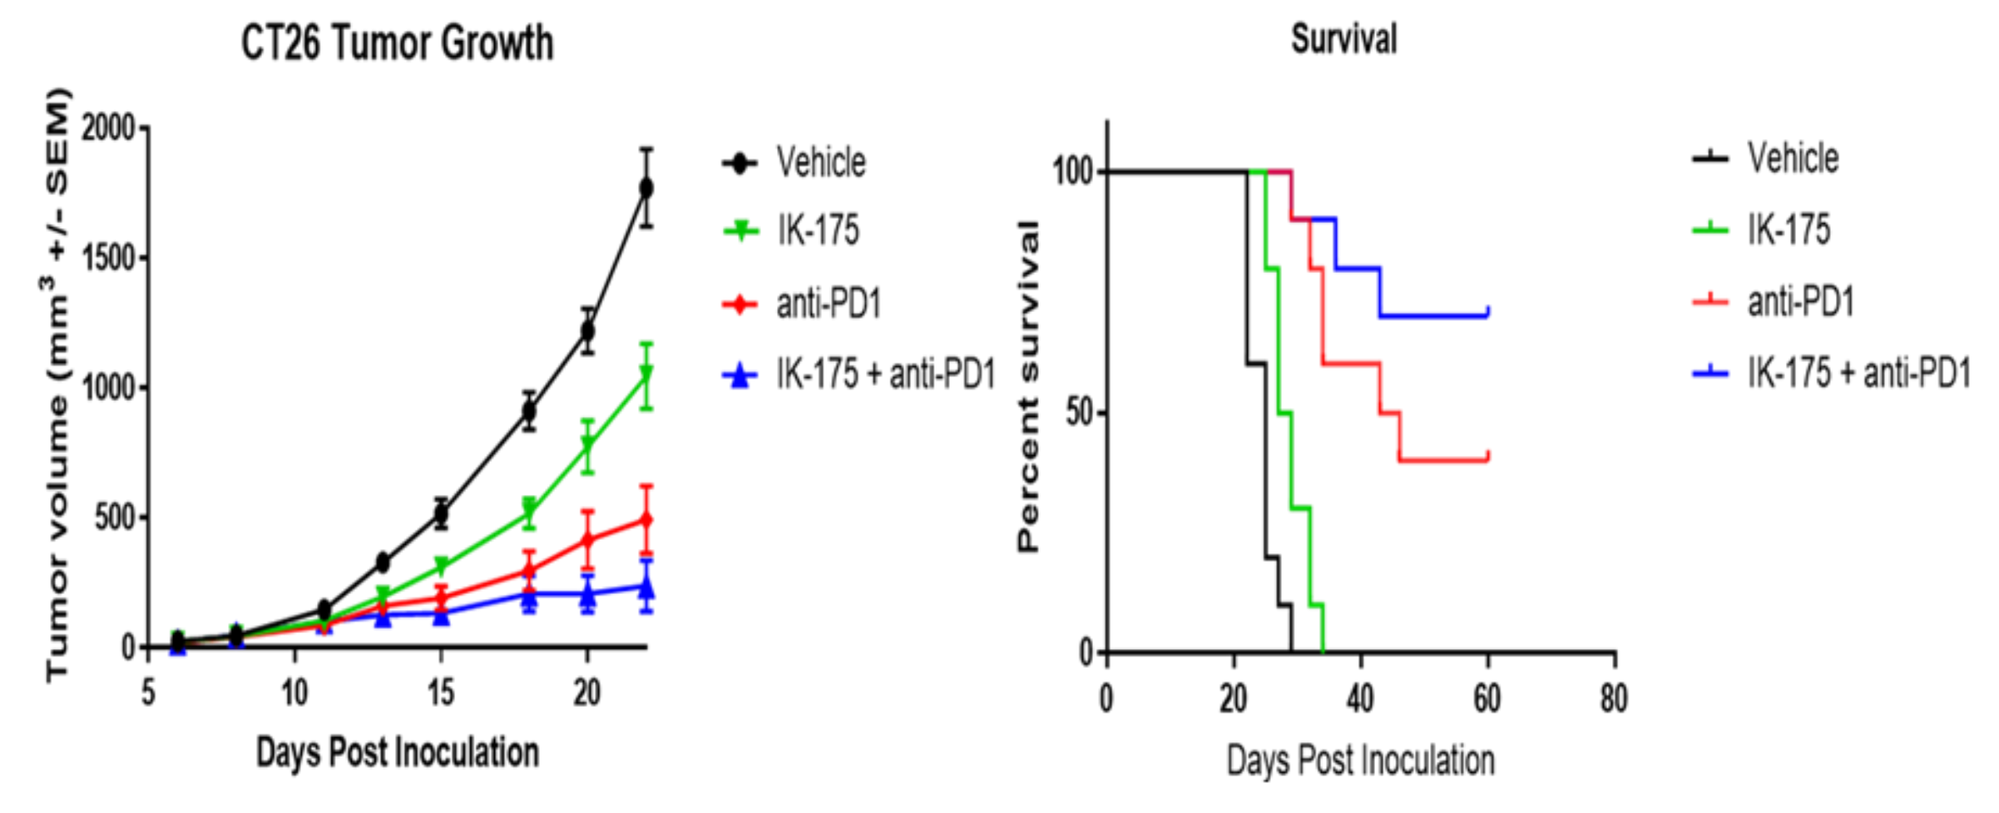
CT26 Tumor Growth
Tumor volume (mm3 +/- SEM)
Days Post Inoculation
Vehicle
IK-175
anti-PD1
IK-175 + anti-PD1
Survival
Percent survival
Days Post Inoculation

IK-175 Single Agent and Combination activity with anti-PD-1 on CT26 tumor growth. IK-175 (25mg/kg, PO, QD); anti-PD-1 (10mg/kg, IP, BIW, 5 doses). p=0.0015 for IK-175 vs. vehicle

- **There were 4 complete responses (CRs) in the anti-PD-1 group and 7 CRs in the IK-175 and anti-PD-1 groups**
- **Upon re-challenge with CT26 there was no tumor growth in the CR mice**
- **Combination group had improved survival compared to Single Agent groups**
- **Similar activity was demonstrated in the B16-IDO orthotopic model**

STUDY DESIGN

- First-in-human (FIH); phase 1a/b open-label, multicenter, dose escalation and expansion study to evaluate the safety, tolerability, pharmacokinetics (PK), pharmacodynamics (PD), and preliminary antitumor activity of **IK-175 dosed as a single agent and in combination with nivolumab in patients diagnosed with locally advanced or metastatic solid tumors and urothelial carcinoma**
- Treatment schedule is administered in 28-day cycle for both treatment arms; For the combination arm, **nivolumab is administered at a fixed dose of 480 mg on day 1 of 28 days cycle** (q4w)
- **Prescreening assessment for AHR nuclear localization by IHC as a predictive biomarker in patients with urothelial carcinoma in both treatment arms.**
 - **A minimum of 10 patients having a positive AHR nuclear localization test (cutoff for positive AHR is 65% tumor cells positive for 2+/3+ nuclear AHR by a validated IHC assay**) will be enrolled in the combination arm.
- Subjects continue treatment until disease progression, unacceptable toxicity, or consent withdrawal

STUDY SCHEMAS

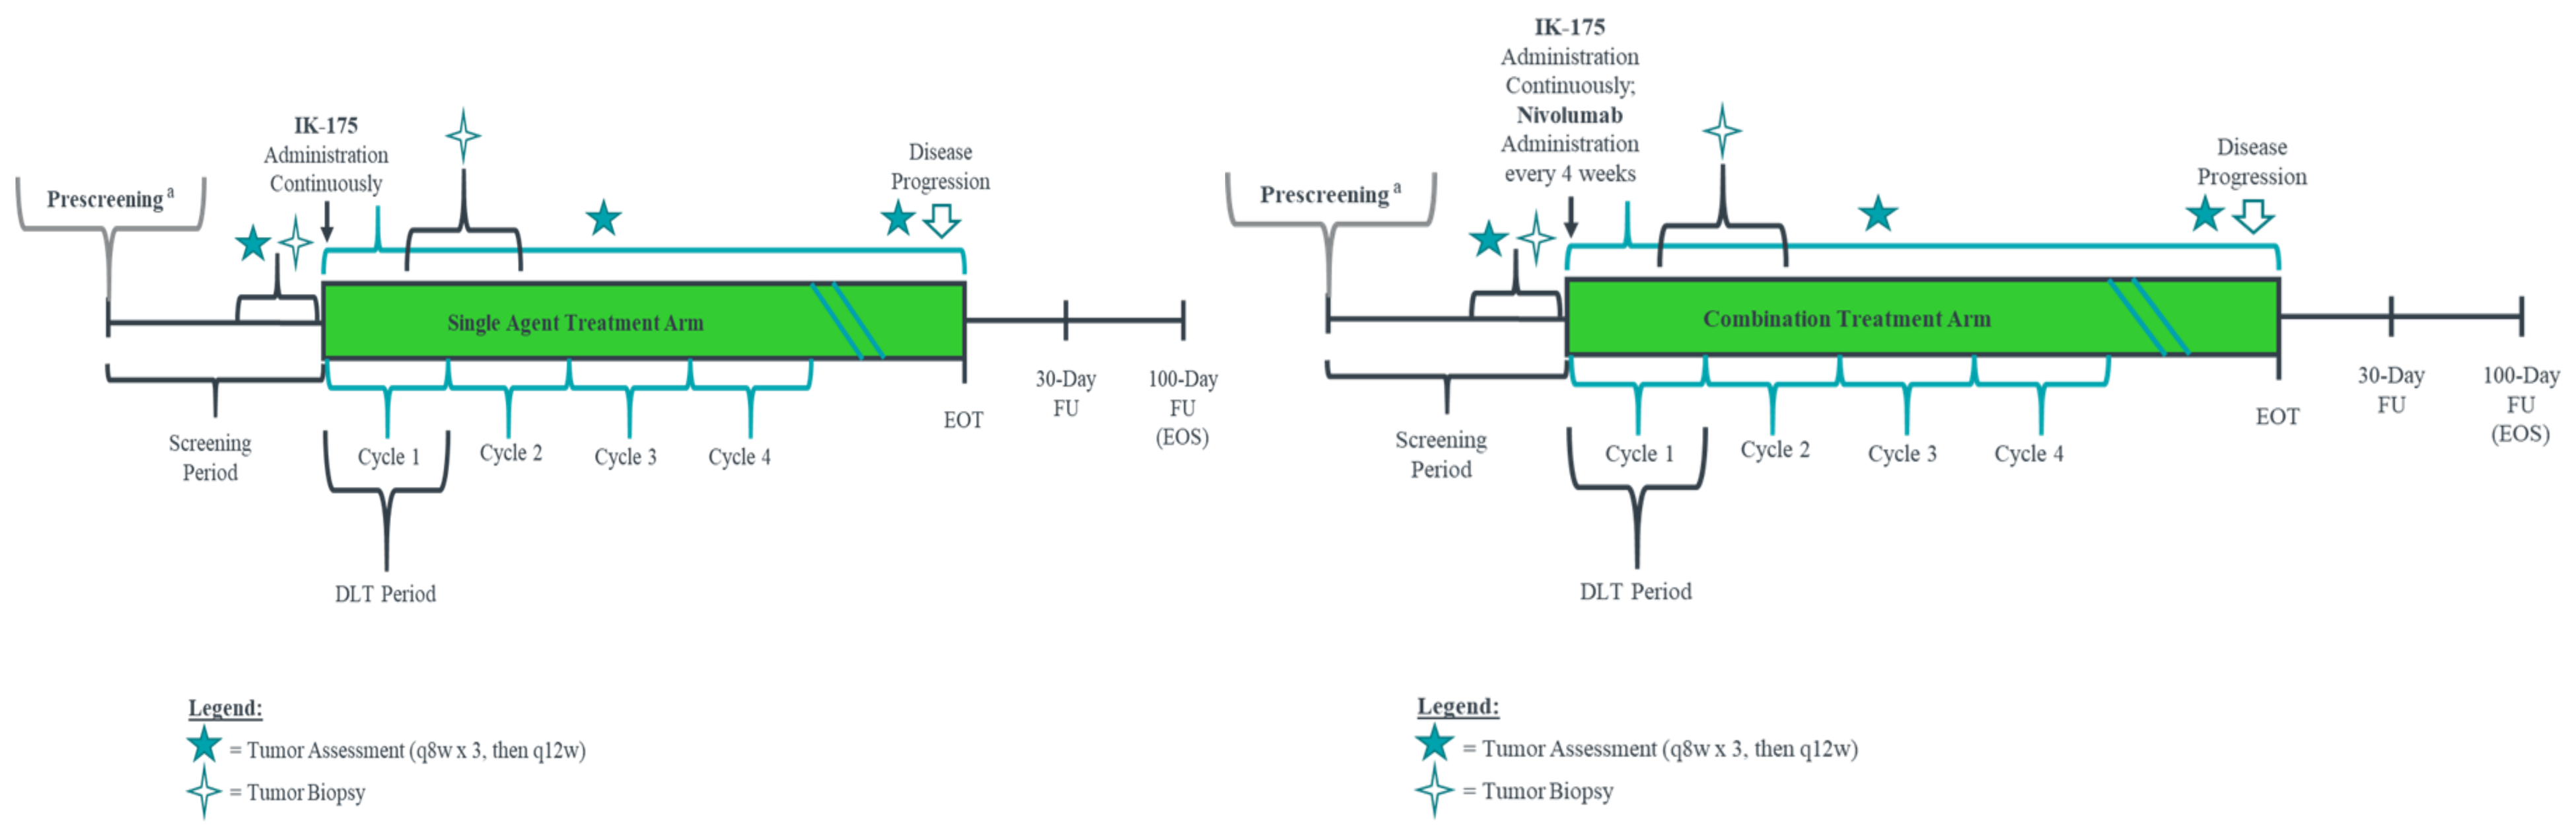
Prescreening a
IK-175 Administration Continuously
Disease Progression
Single Agent Treatment Arm
Screening Period
Cycle 1
Cycle 2
Cycle 3
Cycle 4
EOT
30-Day FU
100-Day FU (EOS)
DLT Period
Legend:
= Tumor Assessment (q8w x 3, then q12w)
= Tumor Biopsy
Prescreening a
IK-175 Administration Continuously;
Nivolumab Administration every 4 weeks
Disease Progression
Combination Treatment Arm
Screening Period
Cycle 1
Cycle 2
Cycle 3
Cycle 4
EOT
30-Day FU
100-Day FU (EOS)
DLT Period
Legend:
= Tumor Assessment (q8w x 3, then q12w)
= Tumor Biopsy

Abbreviations: AHR = aryl hydrocarbon receptor; DLT = dose-limiting toxicity; FU = follow-up; EOS = end of study; EOT = end of treatment; MAD = maximum administered dose; MTD = maximum tolerated dose; q4w = every 4 weeks; q8w = every 8 weeks; q12w = every 12 weeks; QD = once daily.

a. Patients with urothelial carcinoma may consent to a Prescreen AHR nuclear localization assessment and provide archival tumor tissue prior to the Screening period. There is no time limit (i.e., window) for this assessment during the Prescreening period.

KEY ELIGIBILITY CRITERIA

INCLUSION CRITERIA	EXCLUSION CRITERIA
Adult patients with histologically confirmed solid tumors who have locally recurrent or metastatic disease that has progressed on or following all standard of care therapies.	Clinically unstable central nervous system (CNS) tumors or brain metastasis (stable and/or asymptomatic CNS metastases allowed).
Urothelial carcinoma: progressed on or following all standard of care therapies (*e.g.*, platinum-containing regimen and checkpoint inhibitor); combo arm only: progressed on or within 3 months of receiving the last infusion/dose anti-PD-(L)1 therapy.	Any condition requiring continuous systemic treatment with either corticosteroids (>10 mg daily prednisone equivalents) or other immunosuppressive medications within 2 weeks. Inhaled or topical steroids and physiological replacement doses are permitted in the absence of active clinically significant autoimmune disease.
No limit to the number of prior treatment regimens. PD-(L)1 does not need to immediately precede trial.	Active autoimmune disease that has required systemic treatment in past 2 years; nonsteroidal anti-inflammatory drugs (NSAIDs) are permitted.
Have measurable disease per RECIST v1.1	Patients taking strong CYP3A4/5 inhibitors or inducers are excluded from the study unless they can be transferred to other medications ≥ 5 half-lives prior to dosing.

OBJECTIVES & ENDPOINTS

Primary Objectives	Primary Endpoints
To determine the Maximum Tolerated Dose (MTD) or Maximum Administered Dose (MAD), and to characterize the dose limiting toxicities (DLTs) of IK-175 as a single agent and in combination with nivolumab	Proportion of adverse events (AEs) meeting protocol-defined DLT criteria.
To evaluate additional safety and tolerability of IK-175 as a single agent and in combination with nivolumab, including acute and chronic toxicities, in determining a recommended phase 2 dose (RP2D) of IK-175.	Safety endpoint: Frequency of AEs overall, by grade, relationship to study treatment, time-of-onset, duration of the event, duration of resolution, and concomitant medications administered.
Secondary Objectives	**Secondary Endpoints**
To evaluate and characterize the pharmacokinetics (PK) of IK-175 and any major active metabolites	Determination of IK-175 PK parameters, including half-life ($t_{1/2}$), area under the plasma concentration-time curve (AUC) and maximum observed plasma concentration (C_{max}) .
To evaluate disease response with IK-175 treatment as a single agent and in combination with nivolumab.	Preliminary antitumor activity endpoints per RECIST 1.1: Objective response rate (ORR), progression-free survival (PFS), duration of treatment (DOT), disease control rate (DCR), duration of response (DOR). For patients with urothelial carcinoma additional antitumor endpoints include assessment per iRECIST.
To evaluate pharmacodynamic immune effects of IK-175 as a single agent and in combination with nivolumab in collected paired tumor biopsies.	Immune pharmacodynamic endpoints: including but not limited to the tumor infiltrating cytotoxic T cells in tumor biopsies collected before and during IK-175 treatment.
Exploratory Objectives	**Exploratory Endpoints**
To evaluate the PK of IK-175 in a fasting and fed state in a subset of patients.	Summary of IK-175 PK parameters, including t1/2, AUC and Cmax after a single dose in the fasted state, and then after specified doses in the fed state.
To evaluate tumor AHR nuclear localization as a marker associated with disease response to IK-175 as a single agent and in combination with nivolumab.	Association measures of positive AHR nuclear localization assessment with preliminary antitumor activity endpoints per RECIST 1.1 and iRECIST.
To evaluate the pharmacodynamic effects of IK-175 as a single agent and in combination with nivolumab on AHR target gene expression in paired blood draws and paired tumor biopsies.	Changes in AHR target gene expression in blood cells and tumor tissues after study drug treatment.
To evaluate the pharmacodynamic effects of IK-175 as a single agent and in combination with nivolumab on peripheral immune cell and chemokine/cytokine in paired blood draws.	Changes in immune cell types, including but not limited to circulating helper T cells, cytotoxic T cells, and regulatory monocytes after study drug treatment.

STUDY STATUS

The study started in January 2020 and is actively enrolling patients in dose expansion cohorts of both treatment arms. Clinical trial information: NCT04200963

REFERENCES

- McGovern K., Castro A., Cavanaugh J., Discovery of clinical candidate IK-175, a selective orally active AHR antagonist. SITC 2020, poster presentation
- Sanchez-Martin M, Wang L, McGovern K, Ecsedy J and Zhang M.: Computational biology and tissue-based approaches to inform indication selection for a novel AHR inhibitor. SITC 2021, poster presentation (#93)
- Wang L, Sanchez-Martin M, Tirrell S, Ortiz-Otero N and Zhang M.: Analytical validation of a novel immunohistochemistry assay to determine nuclear AHR expression in human bladder cancer . SITC 2021, poster presentation (#58)

ACKNOWLEDGEMENTS

- Ikena Oncology would like to recognize and express gratitude to Dr Jason Sager for his contributions to the implementation of this trial.